Poster Sa1916

REAL WORLD OUTCOMES OF A NOVEL FECAL MICROBIOME REPLACEMENT TREATMENT FOR THE PREVENTION OF RECURRENT *CLOSTRIDIOIDES DIFFICILE* INFECTION: A MULTICENTER STUDY

Timothy E. Ritter, MD[1]; Jonathan A. Rosenberg, MD[2]; Richard L. Hengel, MD[3]; Sujatha Krishnan, MD[4]; Kathy A. Baker, PhD, APRN[5]; Lucinda J. Van Anglen, PharmD[6]; Kelly E. Hanna, PharmD[6]; Amy Guo, PhD[7]; Mielad Moosapanah, PharmD[7]; Kevin W. Garey, PharmD, MS[8]

[1]GI Alliance, Southlake, Texas; [2]GI Alliance, Highland Park, IL; [3]Atlanta ID Group, Atlanta, GA; [4]DFW Infectious Disease, Dallas, TX; [5]Harris College of Nursing and Heath Sciences, Texas Christian University, Fort Worth, Texas; [6]Healix Infusion Therapy, LLC, Sugar Land, Texas; [7]Ferring Pharmaceuticals, Parsippany, NJ; [8]University of Houston College of Pharmacy, Houston, Texas

Introduction

Fecal microbiota, live-jslm (RBL) is the first FDA-approved live biotherapeutic for the prevention of recurrence of *Clostridioides difficile* infection (rCDI) in adults.[1] It is a rectally administered, pre-packaged single dose microbiome therapy, demonstrated to be safe and efficacious in clinical trials.[2,3] This is the first real-world study reporting outcomes of RBL in the US.

Objective

The objective of this study is to evaluate real world outcomes of RBL in prevention of rCDI when used in routine clinical practice.

Methods

- Study design: retrospective multicenter single-arm study
- Data source: electronic health records from February 2023 through November 2023
- Patient population: rCDI patients ≥18 years old who received RBL among 16 separate gastroenterology or infectious disease physician practices in the US
- Study parameters: demographics, comorbidities including Charlson score, number of prior CDI episodes, other prior CDI treatments and rCDI risk factors. Risk factors assessed included age ≥65 years, concurrent gastric acid suppressant use, non-CDI antibiotic use within 4 weeks prior to the current CDI episode, severe presentation of CDI or compromised immune system.
- Utilization characteristics included diagnostic test to confirm CDI, standard of care (SoC) antibiotic used to treat CDI with duration, time from completion of SoC antibiotic to treatment with RBL
- Efficacy assessment: effectiveness of RBL was assessed as absence of recurrence at 8 weeks after treatment. Recurrence was defined as 3 or more liquid bowel movements within 24 hours that required CDI-related therapy.
- Statistical analysis: continuous data are reported as means with standard deviations (SDs) or medians with interquartile ranges (IQRs) and categorical data as counts and percentages. Risk factors for rCDI were calculated with two-tailed Fishers Exact test or Wilcoxon Rank Sum test.

Study Design

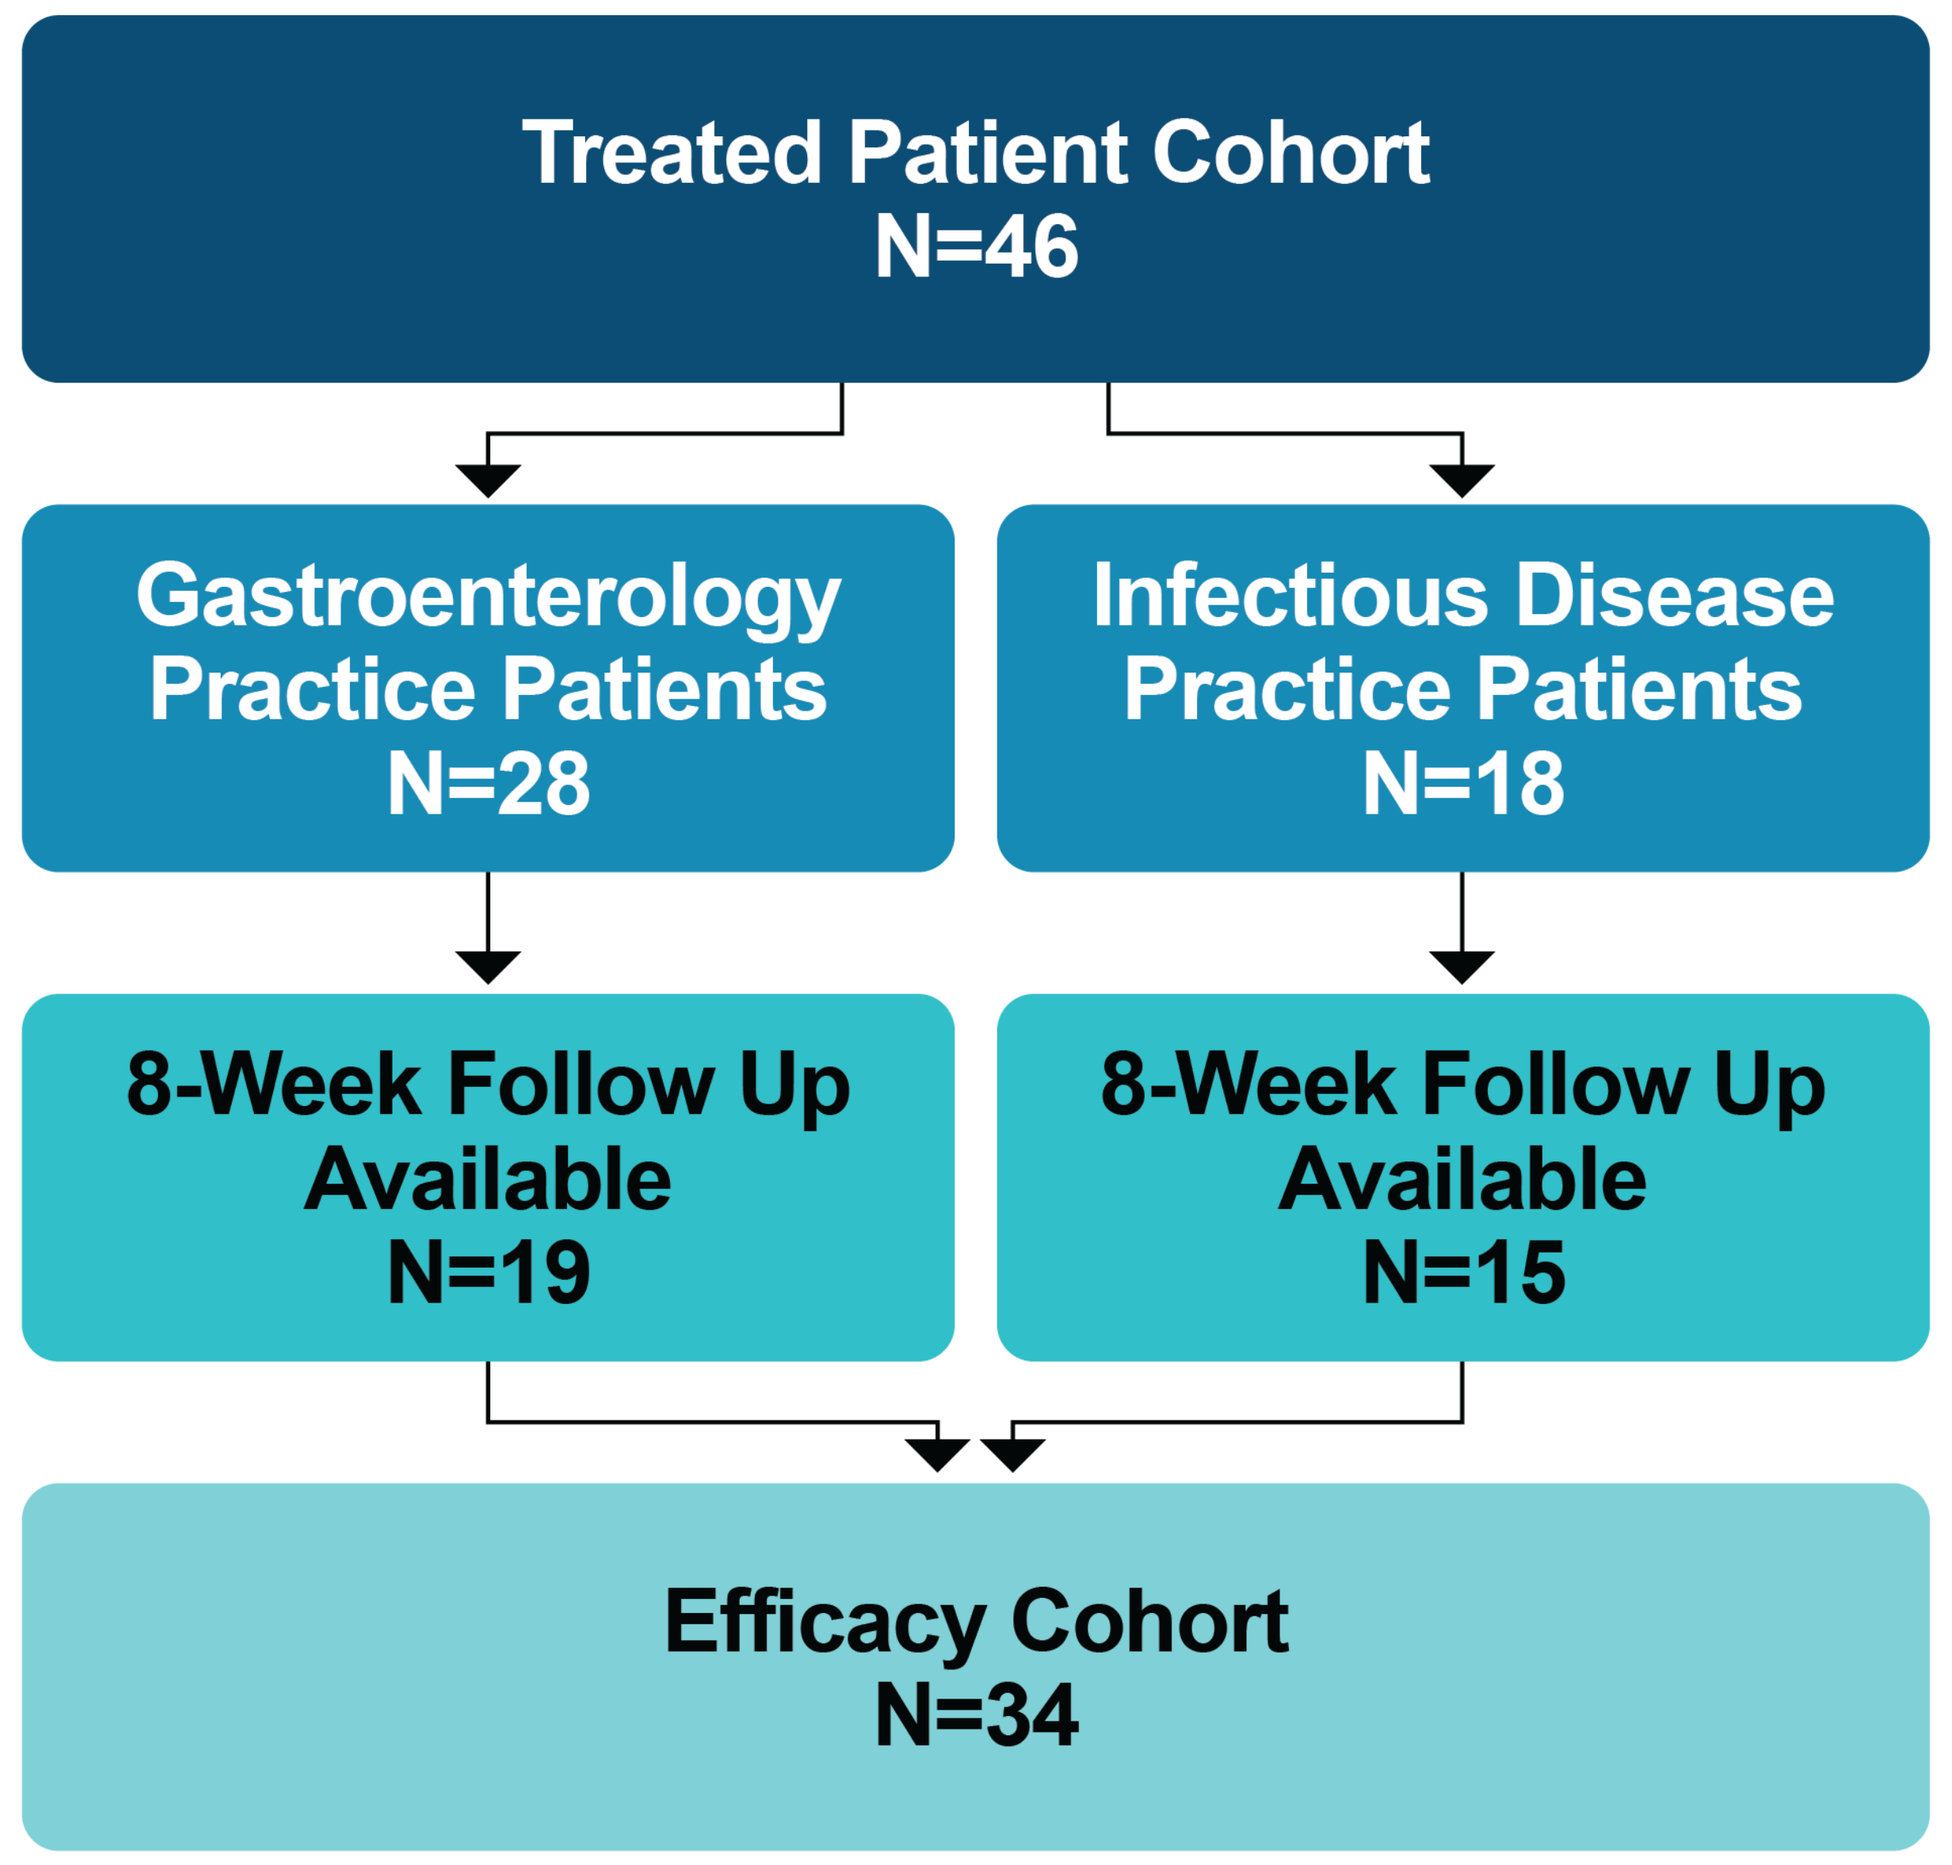

Treated Patient Cohort
N=46
Gastroenterology Practice Patients
N=28
Infectious Disease Practice Patients
N=18
8-Week Follow Up Available
N=19
8-Week Follow Up Available
N=15
Efficacy Cohort
N=34

Results

Baseline Demographics and Characteristics

Characteristic	Results (N=34)
Age, years	
Mean (SD)	70.2 (15.4)
Median (IQR)	75 (64.8-81)
Female	21 (61.8)
Hospitalization within 4 weeks of current CDI	2 (5.9)
Charlson comorbidity index, median (IQR)	4.5 (3-6.8)
CDI history	
Number of prior episodes, not including current, median (IQR)	4 (3-4)
1 episode	5 (14.7)
2 episodes	5 (14.7)
≥3 episodes	24 (70.6)
rCDI risk factors	
Age ≥65 years	25 (73.5)
Concurrent gastric acid suppressant use[a]	20 (58.8)
Non-CDI antibiotic use within 4 weeks prior to current CDI	9 (26.5)
Current CDI with severe presentation[b]	5 (14.7)
Immunocompromised[c]	7 (20.6)
Diagnostic Test Method for *C. difficile* confirmation	
PCR	10 (29.4)
EIA for toxin A, B	6 (17.6)
EIA for toxin A, B + PCR	5 (14.7)
PCR + GDH antigen	5 (14.7)
EIA for toxin A, B + GDH antigen	1 (2.9)
Other characteristics	
Prior therapy with bezlotoxumab	5 (14.7)
Prior FMT	0 (0)
Inflammatory bowel disease	1 (2.9)

Data are presented as number of patients (%) unless otherwise indicated. Abbreviations: EIA, enzyme immunoassay; FMT, fecal microbiota transplant; GDH, glutamate dehydrogenase; PCR, polymerase chain reaction.
[a]Proton pump inhibitor and histamine-2 receptor antagonist.
[b]Defined by any of the following: albumin ≤3.0 g/dl, serum creatinine ≥1.5 times above baseline, hypotension or shock, intensive care unit stay related to CDI, ileus, serum lactate >5 mmol/L, toxic megacolon or colectomy related to CDI, white blood cell count ≥15,000 cells/mL
[c]Due to immunosuppressive medication or underlying disease (immune deficiency, solid organ or hematopoietic stem cell transplant, absolute neutrophil cell count <500 cells/mL).

- Median age was 75 years with a majority female.
- The population was highly comorbid with a median Charlson score of 4.5.
- Over 70% of patients had ≥3 prior CDI episodes.
- Age ≥65 years was the most predominant risk factor, followed by gastric acid suppressant use and non-CDI antibiotics.

rCDI Treatment and RBL Utilization

Characteristic	Results (N=34)
SoC antibiotic with current episode[a]	
Fidaxomicin	17 (50.0)
Vancomycin fixed dose	15 (44.1)
Vancomycin tapered dose	5 (14.7)
Metronidazole	1 (2.9)
RBL utilization parameters	
Days from *C. difficile* stool test to RBL, median (IQR)	36 (25-65)
Days from completion of SoC to RBL, median (IQR)	2 (1.25-4)

[a]Three patients received two SoC antibiotics

Median Duration of SoC Antibiotic

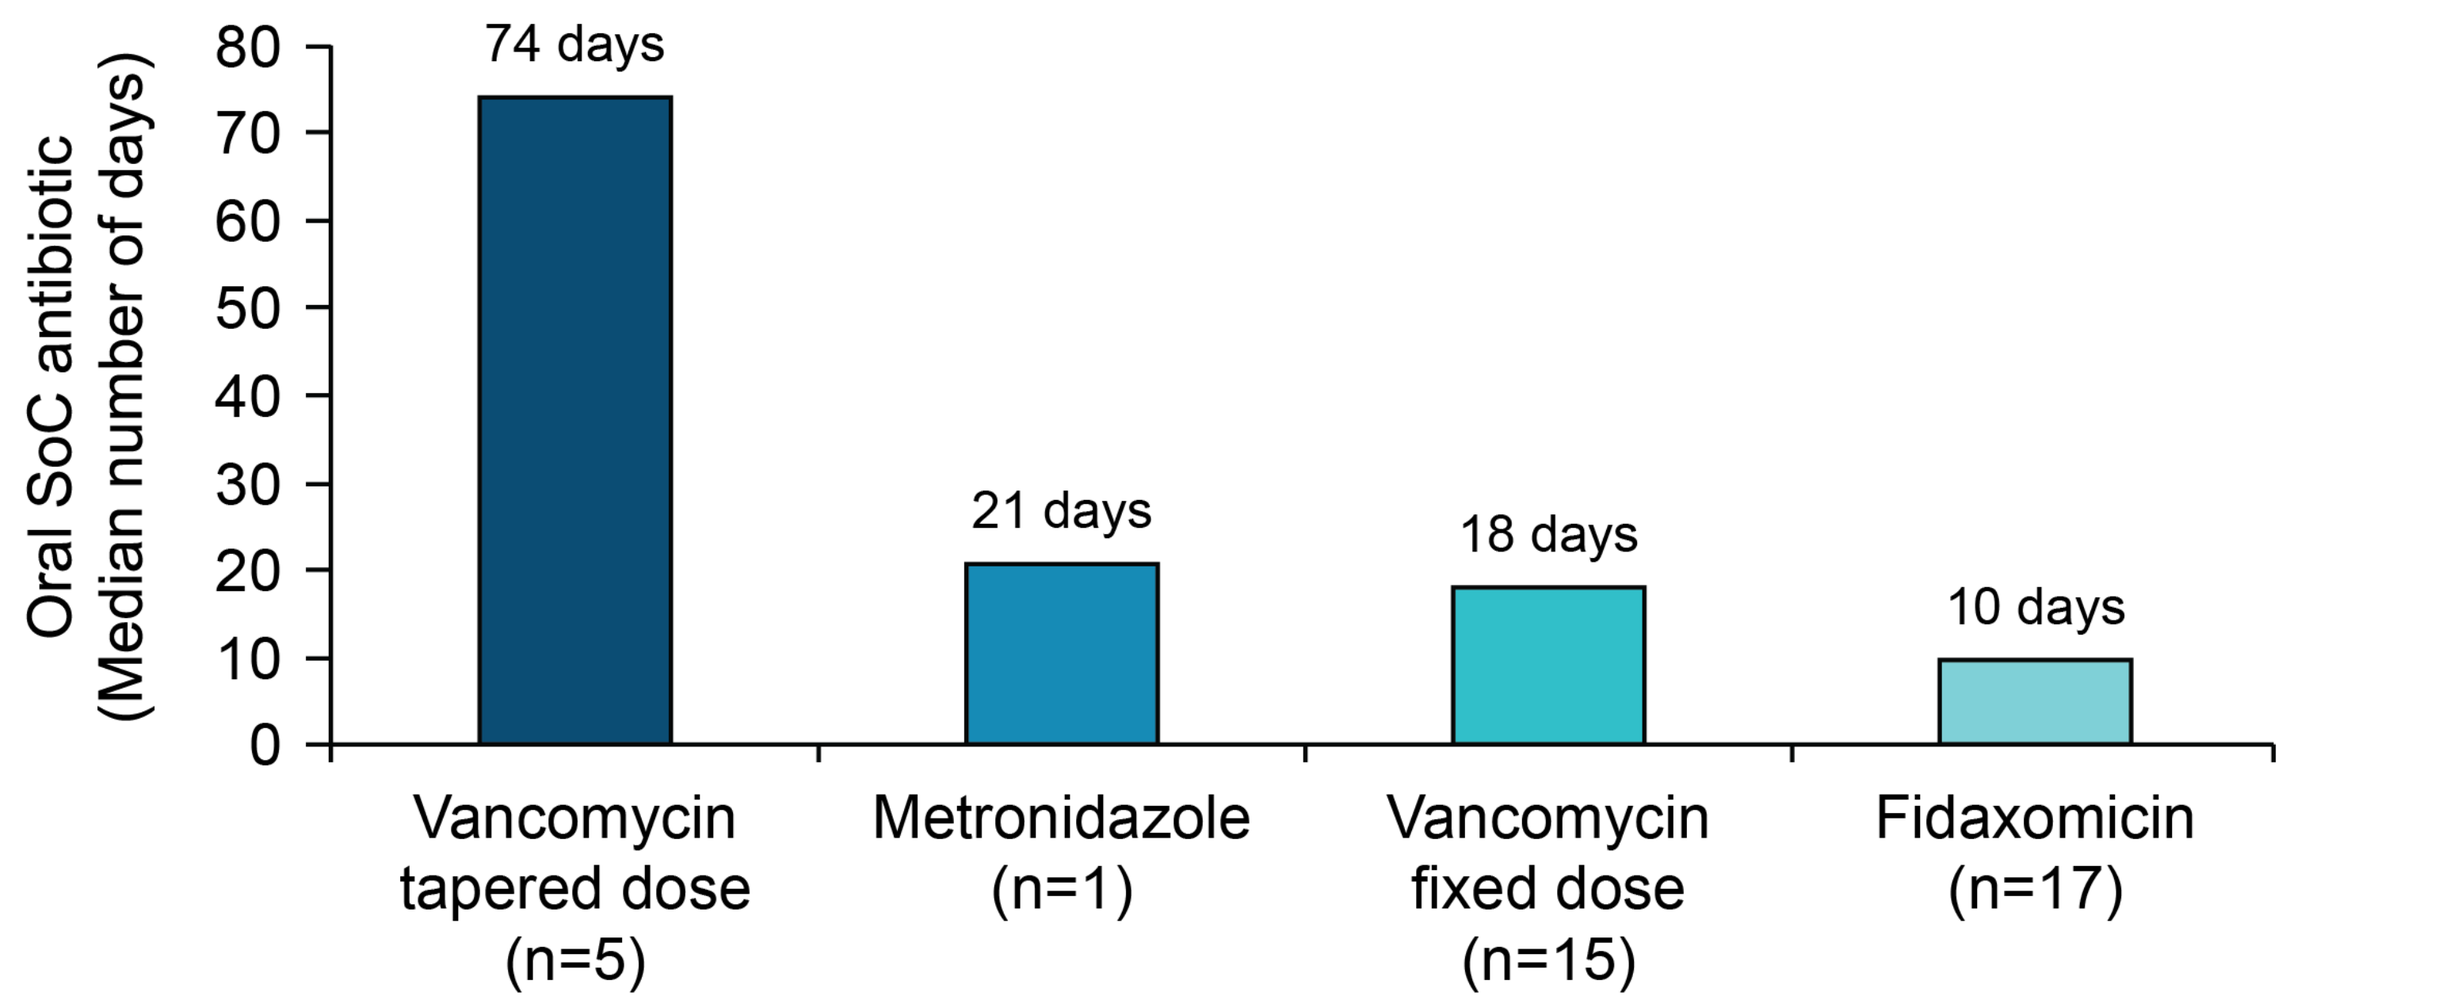

Oral SoC antibiotic (Median number of days)
80
70
60
50
40
30
20
10
0
74 days
21 days
18 days
10 days
Vancomycin tapered dose (n=5)
Metronidazole (n=1)
Vancomycin fixed dose (n=15)
Fidaxomicin (n=17)

Recurrence Rate at 8 Weeks post-RBL

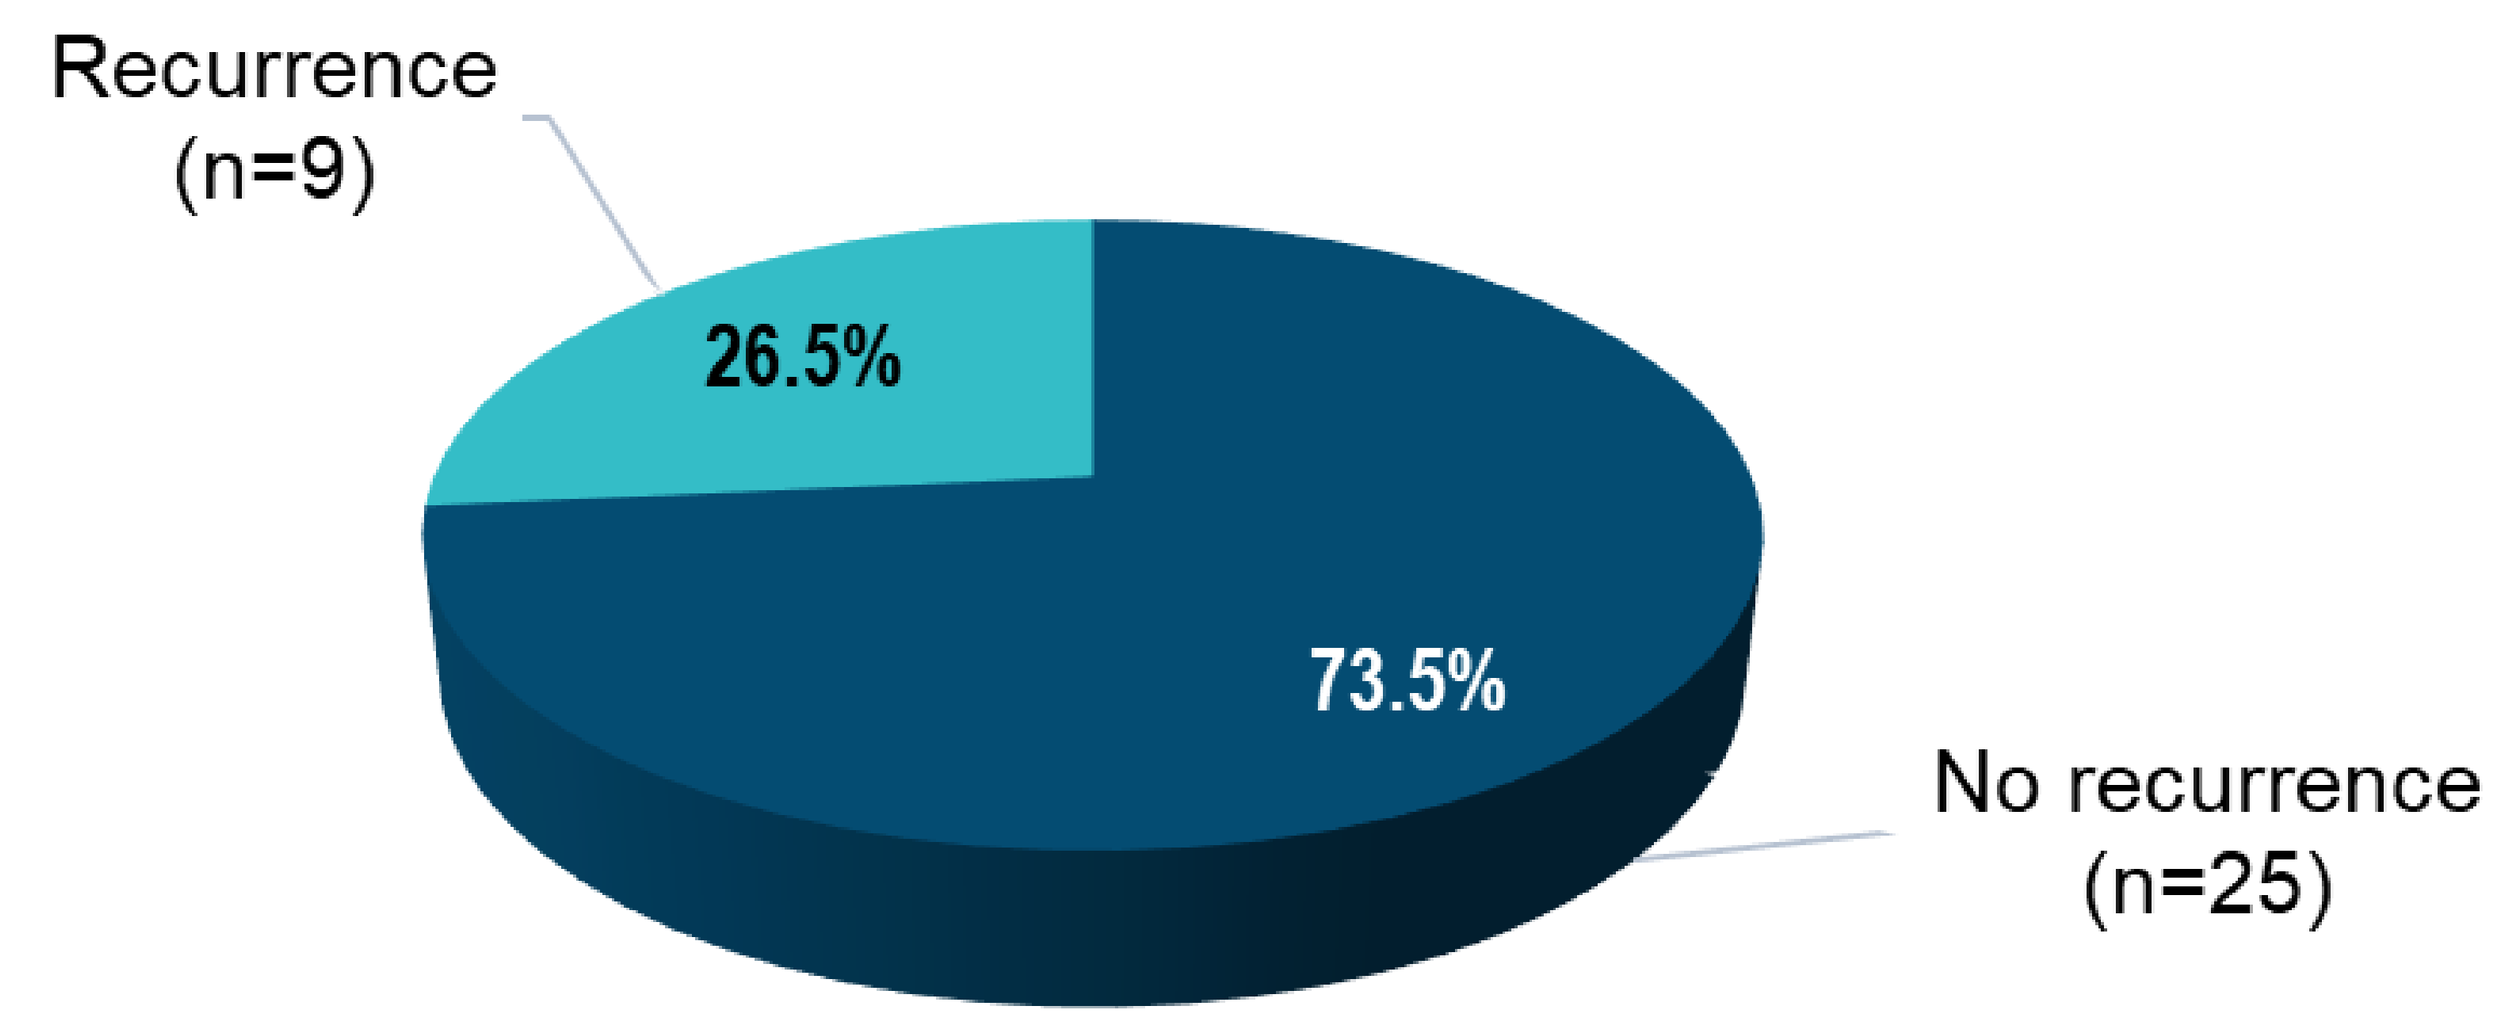

Recurrence (n=9)
26.5%
73.5%
No recurrence (n=25)

- Of the 9 patients who recurred, none were hospitalized for the recurrence.
- One patient with recurrence was re-treated with RBL 20 days following rCDI with no subsequent recurrence at 8 weeks.
- One patient who had inflammatory bowel disease did not have a recurrence of CDI.

Distribution of Recurrence over Time

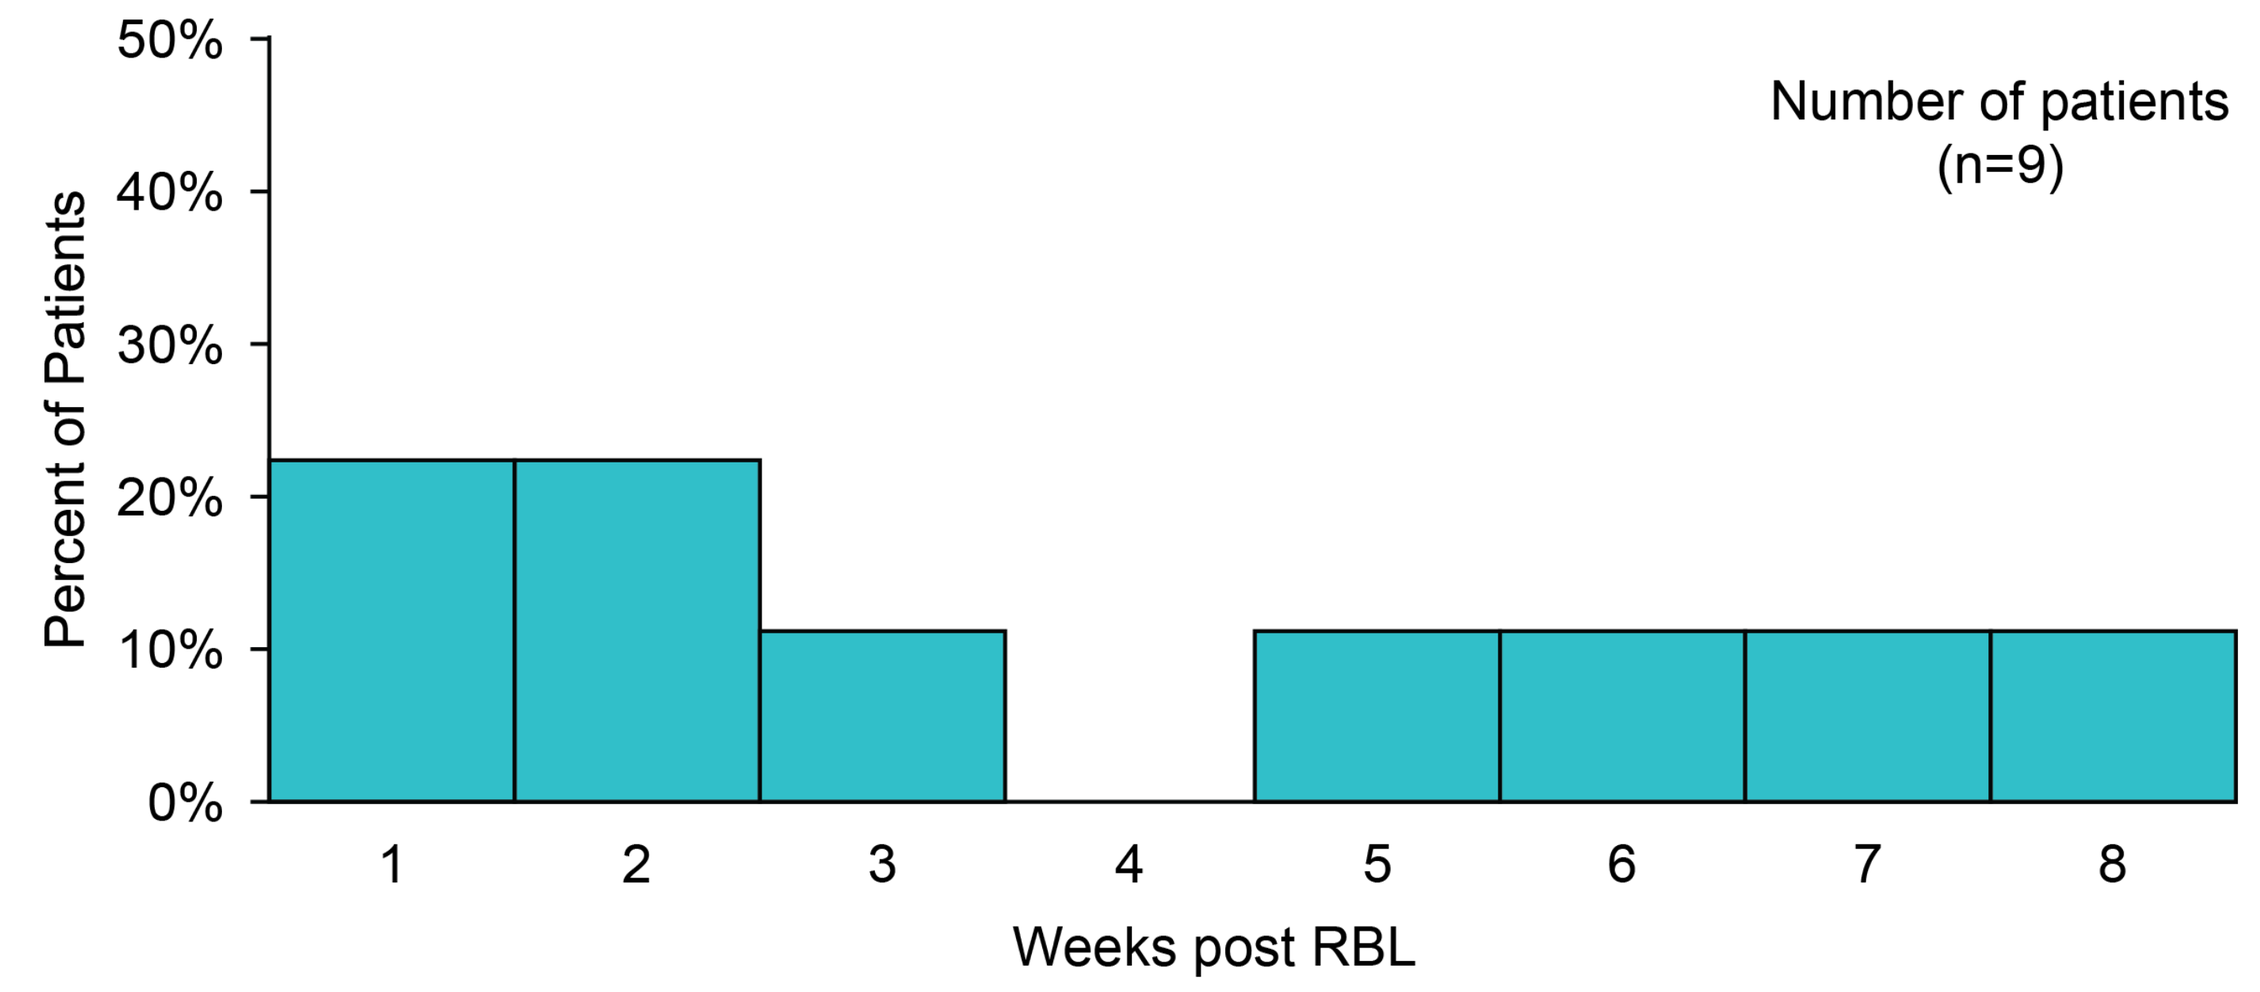

Number of patients (n=9)
Percent of Patients
50%
40%
30%
20%
10%
0%
1
2
3
4
5
6
7
8
Weeks post RBL

Risk Factor Analysis of CDI Recurrence

Parameter (N=34)	No Recurrence (n=25)	Recurrence (n=9)	P-Value[a]
Charlson score, median (IQR)	5 (3-7)	4 (3-6)	0.84
CDI prior episodes, not including current			
1 CDI episode	5 (20%)	0 (0%)	0.29
2 CDI episodes	1 (4%)	4 (44%)	**0.012**
≥3 episodes	19 (76%)	5 (56%)	0.40
Risk factors for rCDI			
Age ≥ 65 years	17 (68%)	8 (89%)	0.39
Gastric acid suppressant	16 (64%)	4 (44%)	0.44
Non-CDI antibiotics within 4 weeks of diagnosis	7 (28%)	2 (22%)	1.00
Immunocompromised	5 (20%)	2 (22%)	1.00
Severe CDI on presentation	4 (16%)	1 (11%)	1.00
Hospitalization within 4 weeks of current CDI	2 (8%)	0 (0%)	1.00
Chronic renal disease	4 (16%)	1 (11%)	1.00
Additional parameters			
Non-CDI antibiotic received post RBL instillation	1 (4%)	1 (11%)	0.47
Inflammatory bowel disease	1 (4%)	0 (0%)	1.00
Treatment with bezlotoxumab for previous episodes	5 (20%)	0 (0%)	0.29
Treatment with bezlotoxumab for current episode	4 (16%)	1 (11%)	1.00
Time from completion of SoC to RBL instillation, median days (IQR)	2 (2-6)	2 (1-2)	**0.035**

[a]P-values were calculated with two-tailed Fishers Exact test or Wilcoxon Rank Sum test

- None of the patients treated after one prior CDI episode experienced a recurrence.
- Patients with 2 prior episodes had a statistically significant reduction in risk of recurrence.
- A longer time from completion of SoC to instillation was associated with a lower risk of recurrence.

Recurrence by CDI Episode

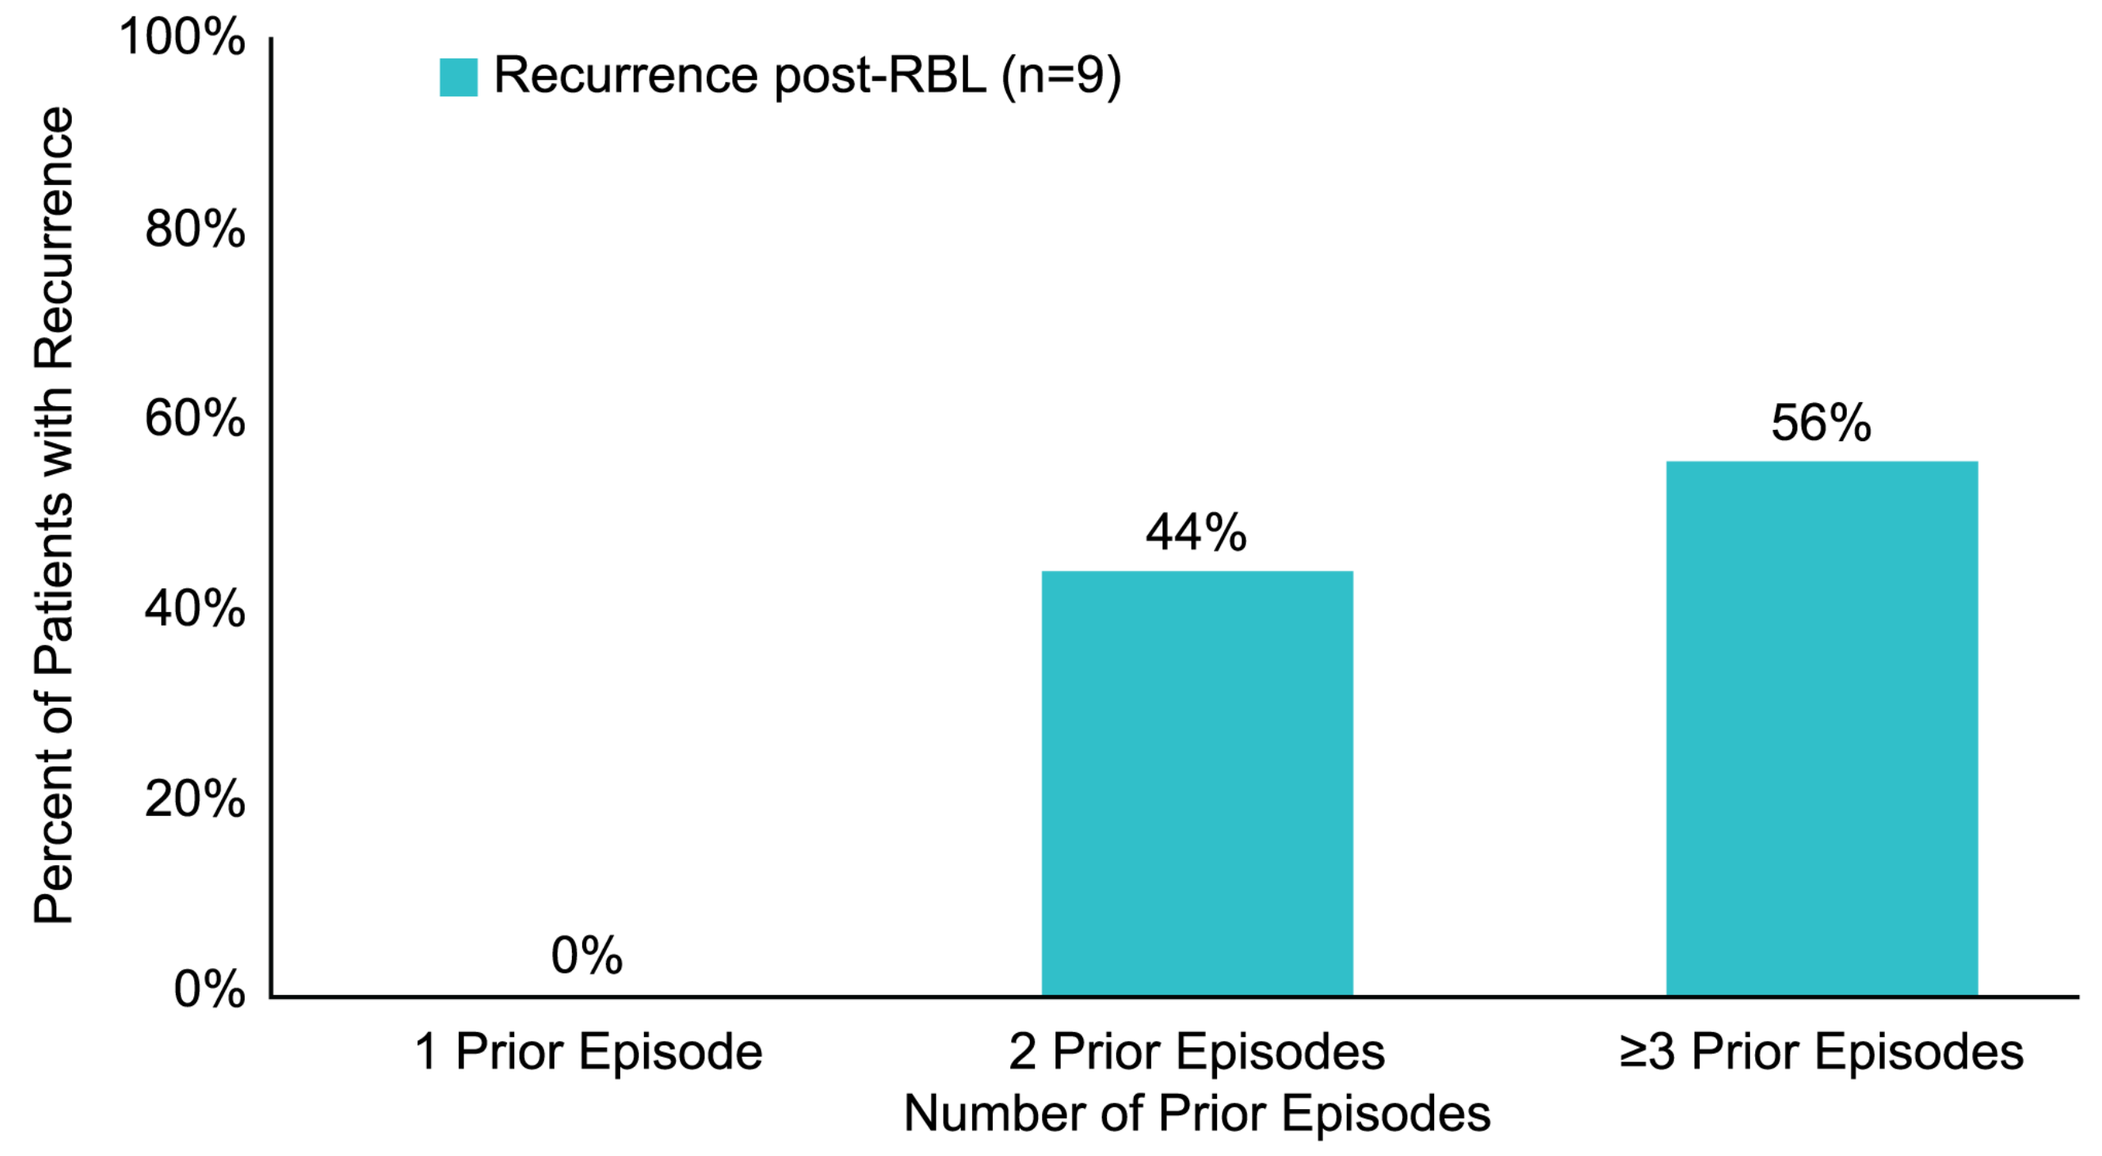

Recurrence post-RBL (n=9)
Percent of Patients with Recurrence
100%
80%
60%
40%
20%
0%
0%
44%
56%
1 Prior Episode
2 Prior Episodes
≥3 Prior Episodes
Number of Prior Episodes

Discussion

This study provides treatment characteristics and efficacy of RBL in prevention of rCDI in a real-world setting.

- 46 patients were treated with RBL in 16 physician offices, with 34 patients evaluated who reached 8 weeks of follow-up.
 - The patient population was highly comorbid with multiple risk factors.
 - Age ≥65 was the most predominant risk factor.
 - Prior CDI episodes were ≥3 in over 70% of patients.
 - Vancomycin was the most used SoC antibiotic.
- Efficacy at 8 weeks with no recurrence of CDI was seen in 73.5%.
- Timing of recurrence was evenly distributed over the 8 weeks.
- None of the patients receiving RBL after one prior CDI episode experienced a recurrence, although this was not significant. Those with two prior episodes did have a significantly lower risk of rCDI.
- No patients who received bezlotoxumab with prior episodes had recurrence following treatment with RBL.
- Patients receiving RBL later after SoC completion had a lower risk of recurrence.
- Limitations include a small sample size for analysis of this interim data and warrants additional study.

Conclusion

- **RBL administered rectally in a physician's office demonstrated 73.5% efficacy in the prevention of recurrence of CDI at 8 weeks.**
- **Patients receiving RBL early had lower rates of recurrence than patients with more prior episodes.**
- **Results are comparable to the data reported in the PUNCH clinical trials, despite a more comorbid patient population with multiple risk factors.[2,3]**

References

1. Rebyota (fecal microbiota, live - jslm) [package insert]. Roseville, MN: Ferring Pharmaceuticals; 2022.
2. Dubberke ER, et al. Infect Dis Ther. 2023; 12:703–710.
3. Khanna S, et al. Drugs. 2022; 82:1527–1538.

This study was funded by a research grant from Ferring Pharmaceuticals Inc.

Digestive Disease Week® 2024 Annual Meeting *May 18-21, 2024; Washington, DC*