IDWeek 2013 #838

Outpatient Parenteral Antimicrobial Therapy (OPAT) for Treatment of Complicated Intra-Abdominal Infections

Infectious Disease Consultants

HEALIX

Fernando S. Alvarado, MD[1], Ramesh V. Nathan, MD[2], H. Barry Baker, MD, FACP[3], Richard L. Levine, MD[3], Andrew H. Krinsky, MD[4], Richard C. Prokesch, MD[5], Phillip S. Brachman, MD[6], Lucinda J. Van Anglen, PharmD[7]

[1]Infectious Disease Consultants, Altamonte Springs, FL, [2]Mazur, Statner, Dutta, Nathan, PC, Thousand Oaks, CA, [3]Infectious Disease Physicians, PA, Miami, FL, [4]Infectious Diseases Associates, Sarasota, FL, [5]Infectious Disease Associates, Riverdale, GA, [6]Atlanta ID Group, PC, Atlanta, GA, [7]Healix Infusion Therapy, Inc., Sugar Land, TX

Lucinda J. Van Anglen, PharmD
Healix Infusion Therapy, Inc.
14140 SW Fwy, 4th Floor
Sugar Land, TX 77478
281-295-4000
Lvananglen@healix.net

Abstract, revised

Background: Optimal treatment of complicated intra-abdominal infections (IAI) requires rapid intervention and initiation of antimicrobial agents. Infectious Disease (ID) physicians with OPAT services offer a closely supervised setting for community and post-hospital treatment of these patients (pts). We report clinical experience of pts treated for IAI with OPAT in a physician office infusion center (POIC).

Methods: A multi-center, retrospective database review of 10 ID POICs was conducted of all pts treated with intravenous antibiotics (IVAB) between January 1 and June 30, 2011. Data included demographics, co-morbidities, infecting organism, drug therapy, adverse events and outcomes. Outcomes were defined as cure, improvement requiring oral antibiotics or further surgical intervention, or failure due to worsening of infection or recurrence requiring IVAB.

Results: 120 pts were identified in 10 POICs, of which 106 (88%) had therapy continued following hospitalization and 14 (12%) had therapy initiated in the POIC. There were 57 (48%) males, and mean age was 56 years. Predominant IAI treated were diverticulitis (34%), abscess (22%), other post-operative abdominal infections (20%), and appendicitis (12%). Pathogens were reported in 72 pts, with 35 polymicrobial infections. Predominant organisms included *E. coli* (21, 18%), *Streptococcus* spp (15, 13%), and *Bacteroides* spp (12, 10%). The most commonly utilized IVAB were piperacillin/tazobactam (30%), ertapenem (17%) and ceftriaxone (12%), with a mean length of therapy of 15 days in the OPAT setting. Cure was reported in 59 (49%) pts and improvement in 45 (38%) pts. 53% of improved cases were discharged with oral therapy and 47% were pending further surgical intervention. 16 (13%) pts failed, 10 of whom had recurrence of infection. A serious adverse event occurred in 1 pt. The catheter infection rate was 0/1000 catheter days.

Conclusion: OPAT appears to be a safe and effective setting for treatment of complicated IAI for pts initiated in the POIC as well as those discharged after hospitalization. Overall rates of cure and improvement were high in this population, with low rates of adverse events and catheter-related infections. Further analysis with long-term follow-up is warranted.

Introduction

Intra-abdominal infections are the second most common cause of infectious mortality in intensive care units in the US. Parenteral antimicrobial therapy is traditionally offered in the inpatient setting; however, an increase in the delivery of OPAT has become an established standard of care.[1]

Complicated intra-abdominal infection, which extends into the peritoneal space, is associated with abscess formation and peritonitis. Uncomplicated infection, which involves intramural inflammation of the gastrointestinal tract, may progress to complicated infection if left untreated.[1] Intra-abdominal infections comprise a wide variety of clinical presentations and differing sources of infection. In fact, a wide variety of bacterial pathogens are responsible for complicated IAI, including gram-negative aerobic bacteria, gram-positive bacteria, anaerobic bacteria, and also mixed infections.[2, 3]

This presentation will review the experience of OPAT in patients with complicated IAIs, discussing antibiotic options and treatment length with respect to specific diagnosis, reported pathogens and the associated clinical outcomes.

Methods

A multi-center, retrospective database review was conducted to identify IAI patients treated with IVABs in POICs nationwide.

Inclusion Criteria:

- IVAB therapy received in POIC between January 1, 2011 and June 30, 2011
- Documented IAI diagnosis

Data Analysis:

- Data from 10 POICs nationwide were collected for patient demographics, diagnoses, selection and duration of drug therapy, their microbiology, adverse events, and clinical outcomes
- Descriptive statistics were used for demographic data
- Creatinine clearance (CrCl) was calculated according to Cockcroft-Gault formula
- Pathogen data were collected where available
- Multidrug resistant (MDR) organisms were defined as organisms resistant to ≥ 3 antimicrobials
- IVAB efficacy at time of POIC discharge was defined as follows:

Cured: Clinical signs/symptoms resolved, and/or no additional antibiotic therapy needed, and/or negative culture at end of therapy

Improved: Partial resolution of clinical signs/symptoms, and/or additional oral antibiotic therapy necessary, and/or wound care or surgical procedure pending

Failed: Resistant, worsening, or new clinical signs/symptoms, and/or recurrence requiring IVAB at therapy completion

Results

Demographics

Table 1. Demographics

Characteristics (n=120)	Patients No. (%)
Gender	
Male	57 (48%)
Age	
Mean Age in Years (range)	56 (20-92)
< 65	87 (73%)
Ethnicity	
Caucasian	96 (80%)
Hispanic	13 (11%)
African American	8 (7%)
Obesity defined as BMI ≥30	43 (36%)
Comorbidities	
Hypertension	58 (48%)
Cardiovascular Disease	25 (21%)
Hypothyroid	21 (18%)
Hyperlipidemia	20 (17%)
Diabetes	14 (12%)
Cancer History	13 (11%)
COPD/Asthma	12 (10%)
Smoker	11 (9%)
Immunocompromised	8 (7%)
CRF/ESRD	6 (5%)
Recent GI Surgery	2 (2%)
Comorbidities per patient	
None	19 (16%)
One	32 (27%)
Two	26 (21%)
Three or more	43 (36%)
Prior Hospitalization	104 (87%)

Diagnoses

Figure 1. Diagnoses

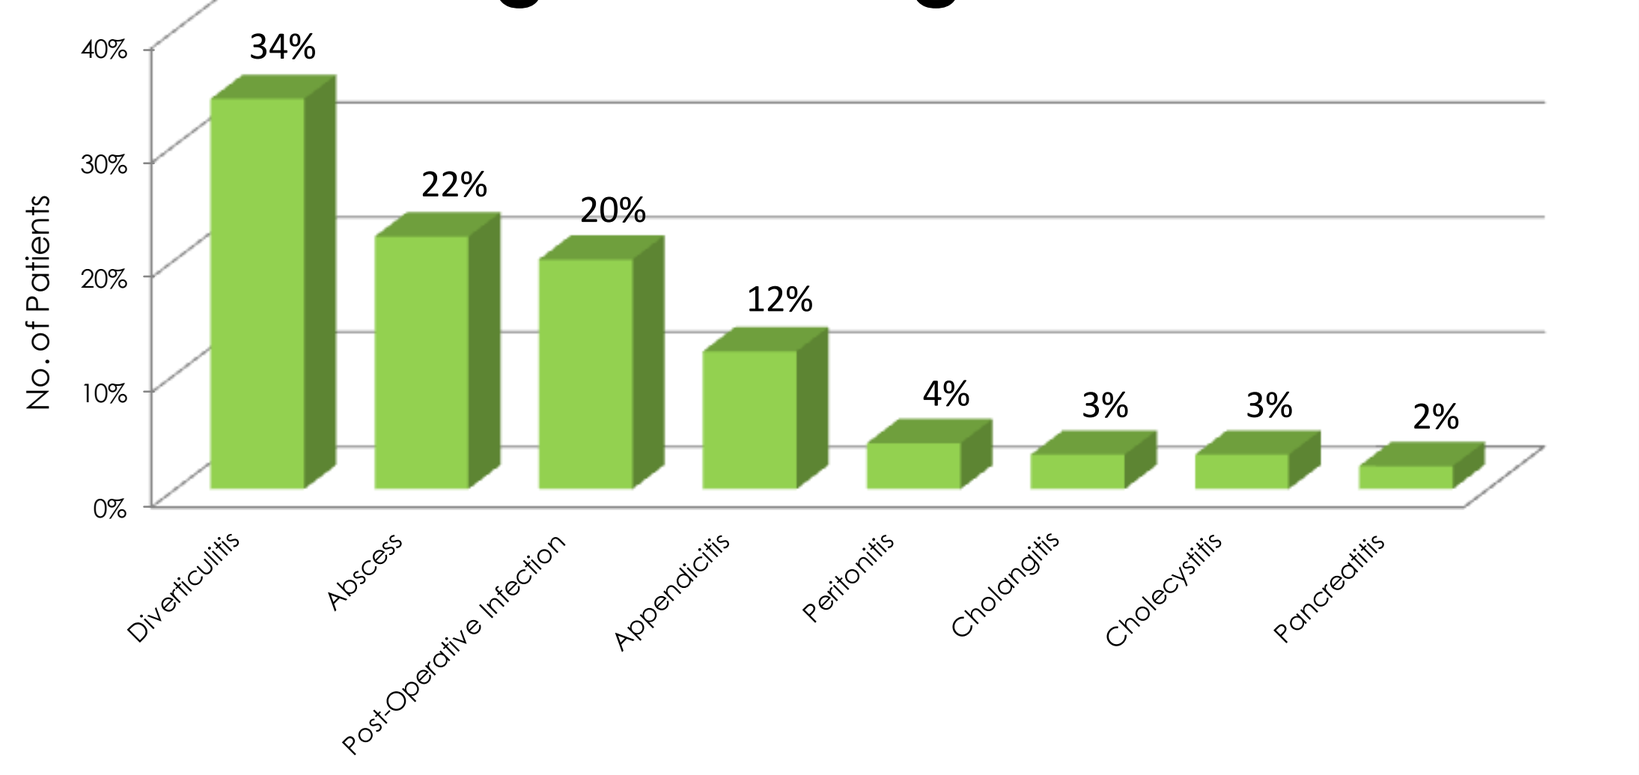
No. of Patients
40%
30%
20%
10%
0%
34%
22%
20%
12%
4%
3%
3%
2%
Diverticulitis
Abscess
Post-Operative Infection
Appendicitis
Peritonitis
Cholangitis
Cholecystitis
Pancreatitis

- Diagnoses were recorded as primary noted by the physician.
- Post-operative infections represented 30% with complications and infections following organ removal, 30% complicated abdominal wall infections from various surgeries, 25% complicated infections following hernia repairs.
- 37% of diverticulitis patients also had abscesses and/or perforations.
- Appendicitis patients were complicated in 73% with abscess, perforation or peritonitis.

Microbiology

Table 2. Incidence of Pathogens in Patients

Organism	Patients No., (%) (n=72)	Reported MDR No., (% resistant organisms)
Facultative and aerobic gram negative		
Eschericia coli	21 (29)	14 (48)
Klebsiella species	7 (10)	7 (100)
Pseudomonas species	7 (10)	2 (29)
Enterobacter species	5 (7)	5 (71)
Citrobacter species	2 (3)	2 (100)
Other gram negative organisms	7 (10)	NR
Anaerobic		
Bacteroides fragilis	6 (8)	1 (17)
Other *Bacteroides* species	6 (8)	NR
Fusobacterium species	3 (4)	NR
Clostridium species	2 (3)	NR
Other anaerobic organisms	2 (3)	NR
Gram-positive aerobic cocci		
Streptococcus species	15 (21)	3 (20)
Staphylococcus aureus	11 (15)	3 (27)
Enterococcus species	7 (10)	4 (57)
Other *Staphylococcus* species	3 (4)	2 (19)
Other gram-positive cocci	4 (6)	NR
Candida species	8 (11)	NR
Mycobacterium chelonae	1 (1)	NR

- Culture data was obtained in 72 patients with 117 pathogens.
- 35 of 72 (49%) patients had polymicrobial infections.
- Resistance was reported in 31 of the 117 (26%) pathogens.

Figure 2. Microbiology by Diagnosis

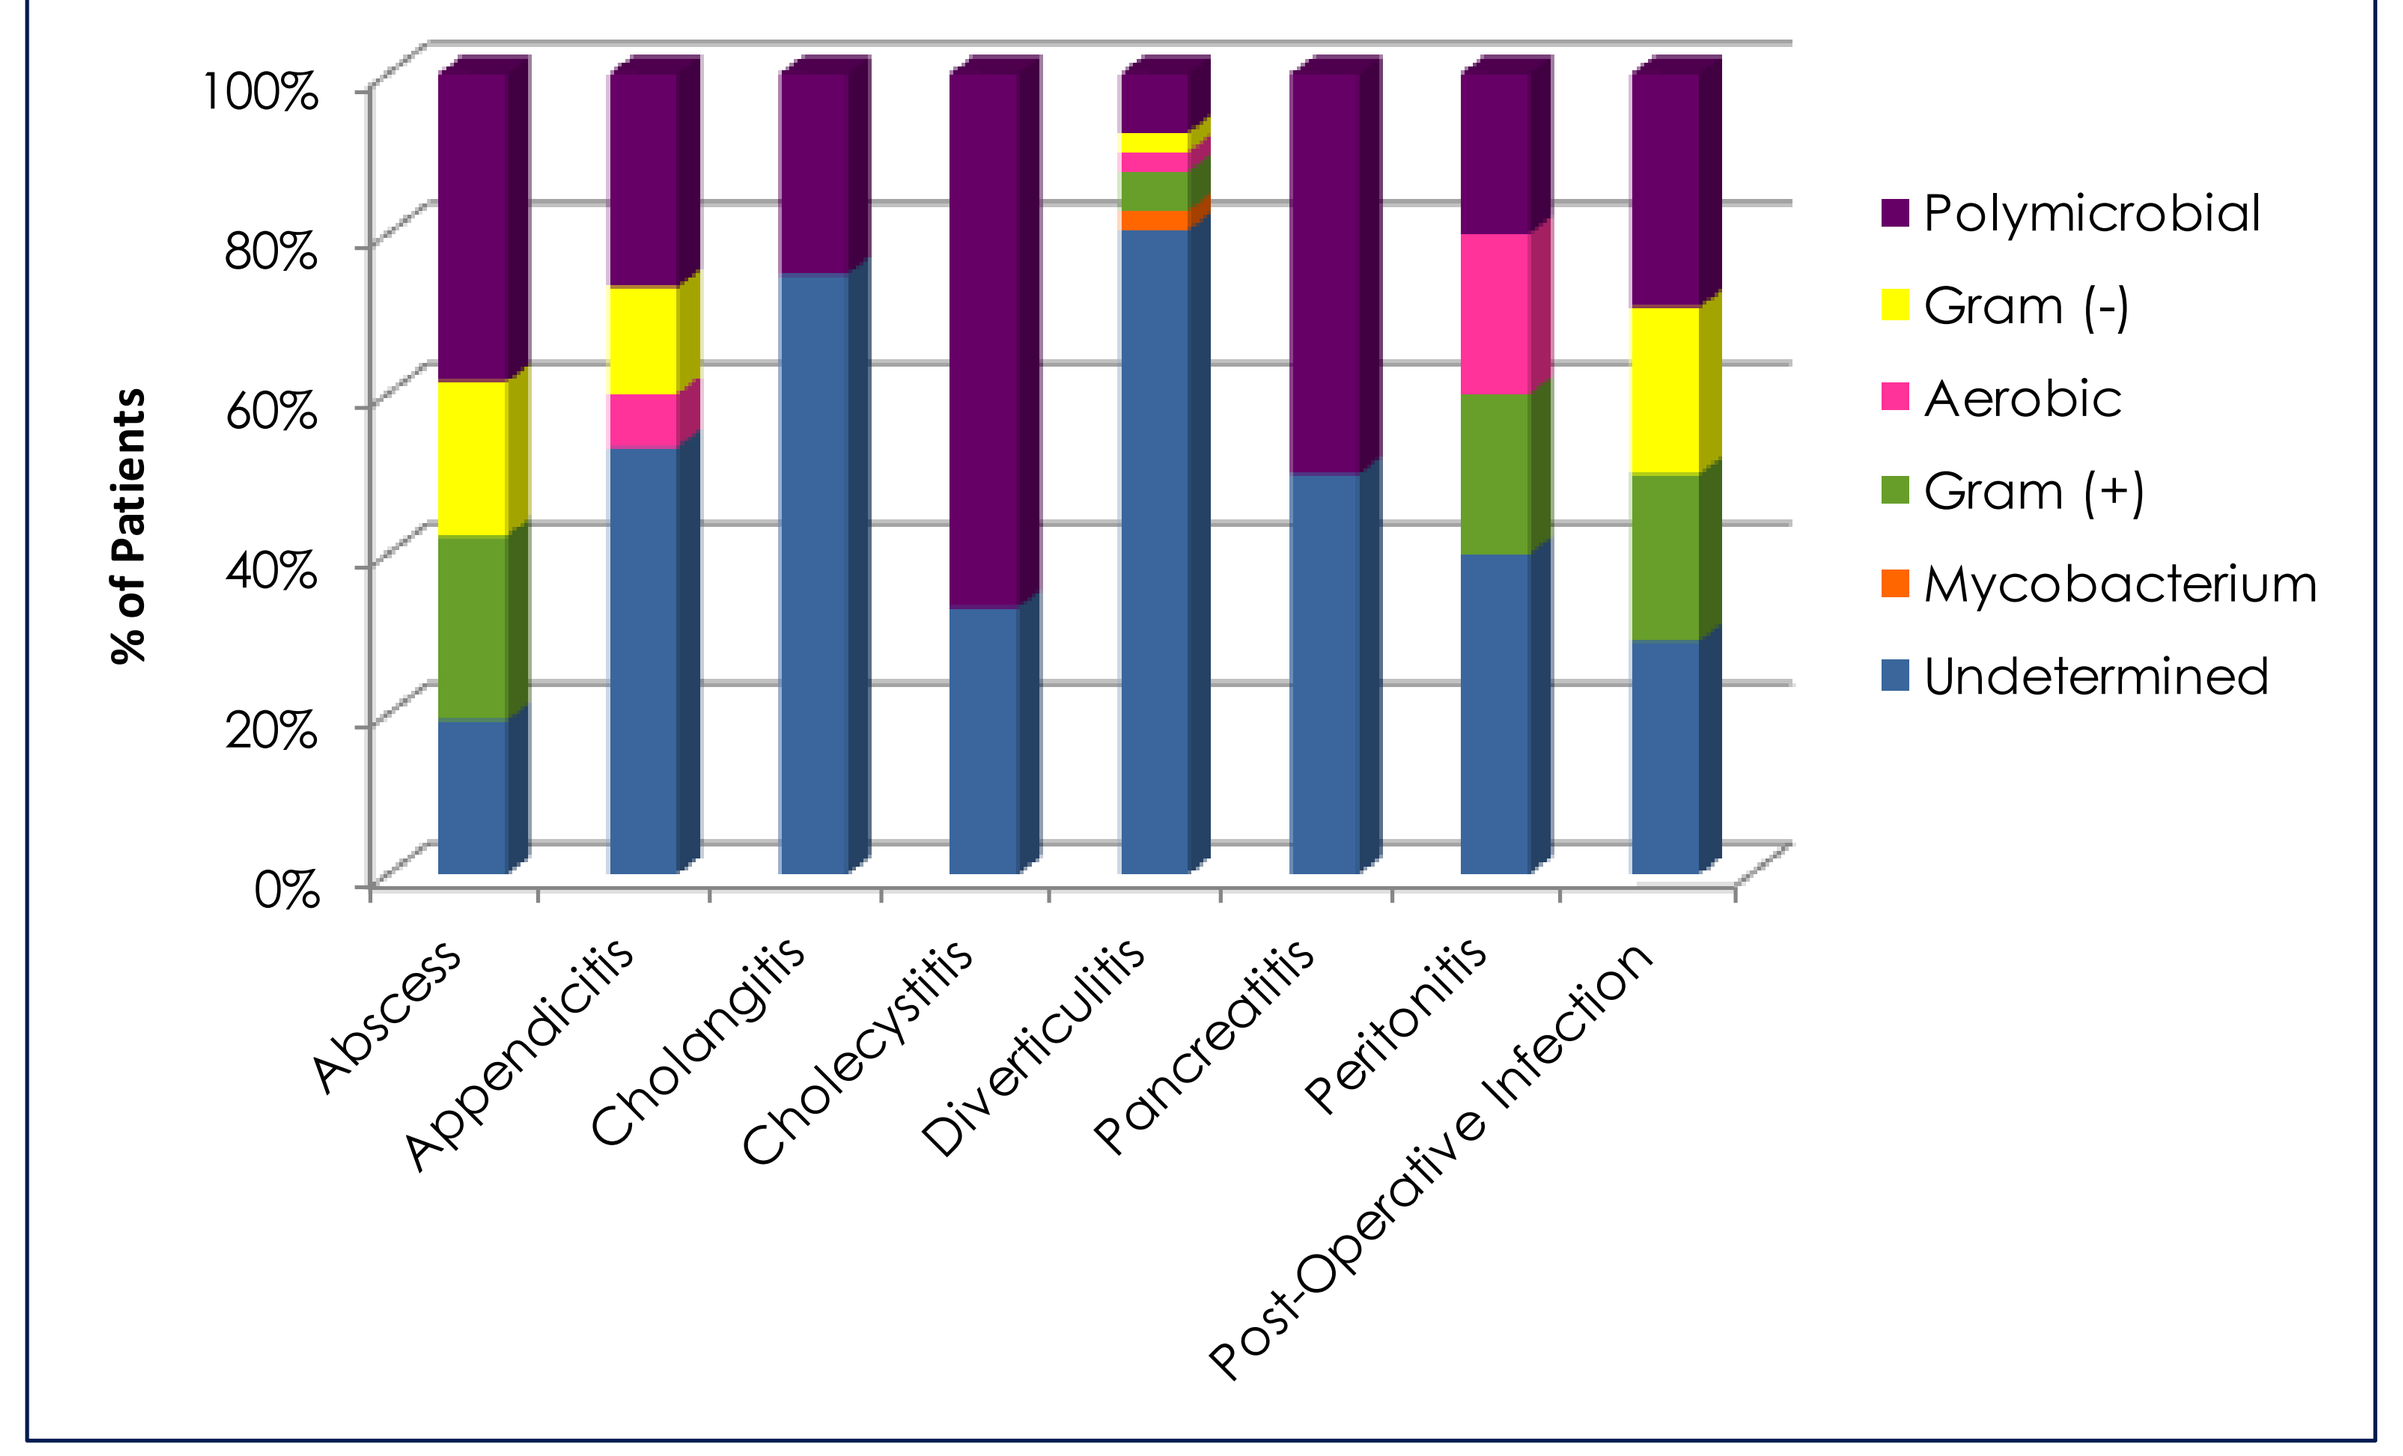
% of Patients
100%
80%
60%
40%
20%
0%
Abscess
Appendicitis
Cholangitis
Cholecystitis
Diverticulitis
Pancreatitis
Peritonitis
Post-Operative Infection
Polymicrobial
Gram (-)
Aerobic
Gram (+)
Mycobacterium
Undetermined

Treatment

Figure 3. Most Commonly Used Drugs

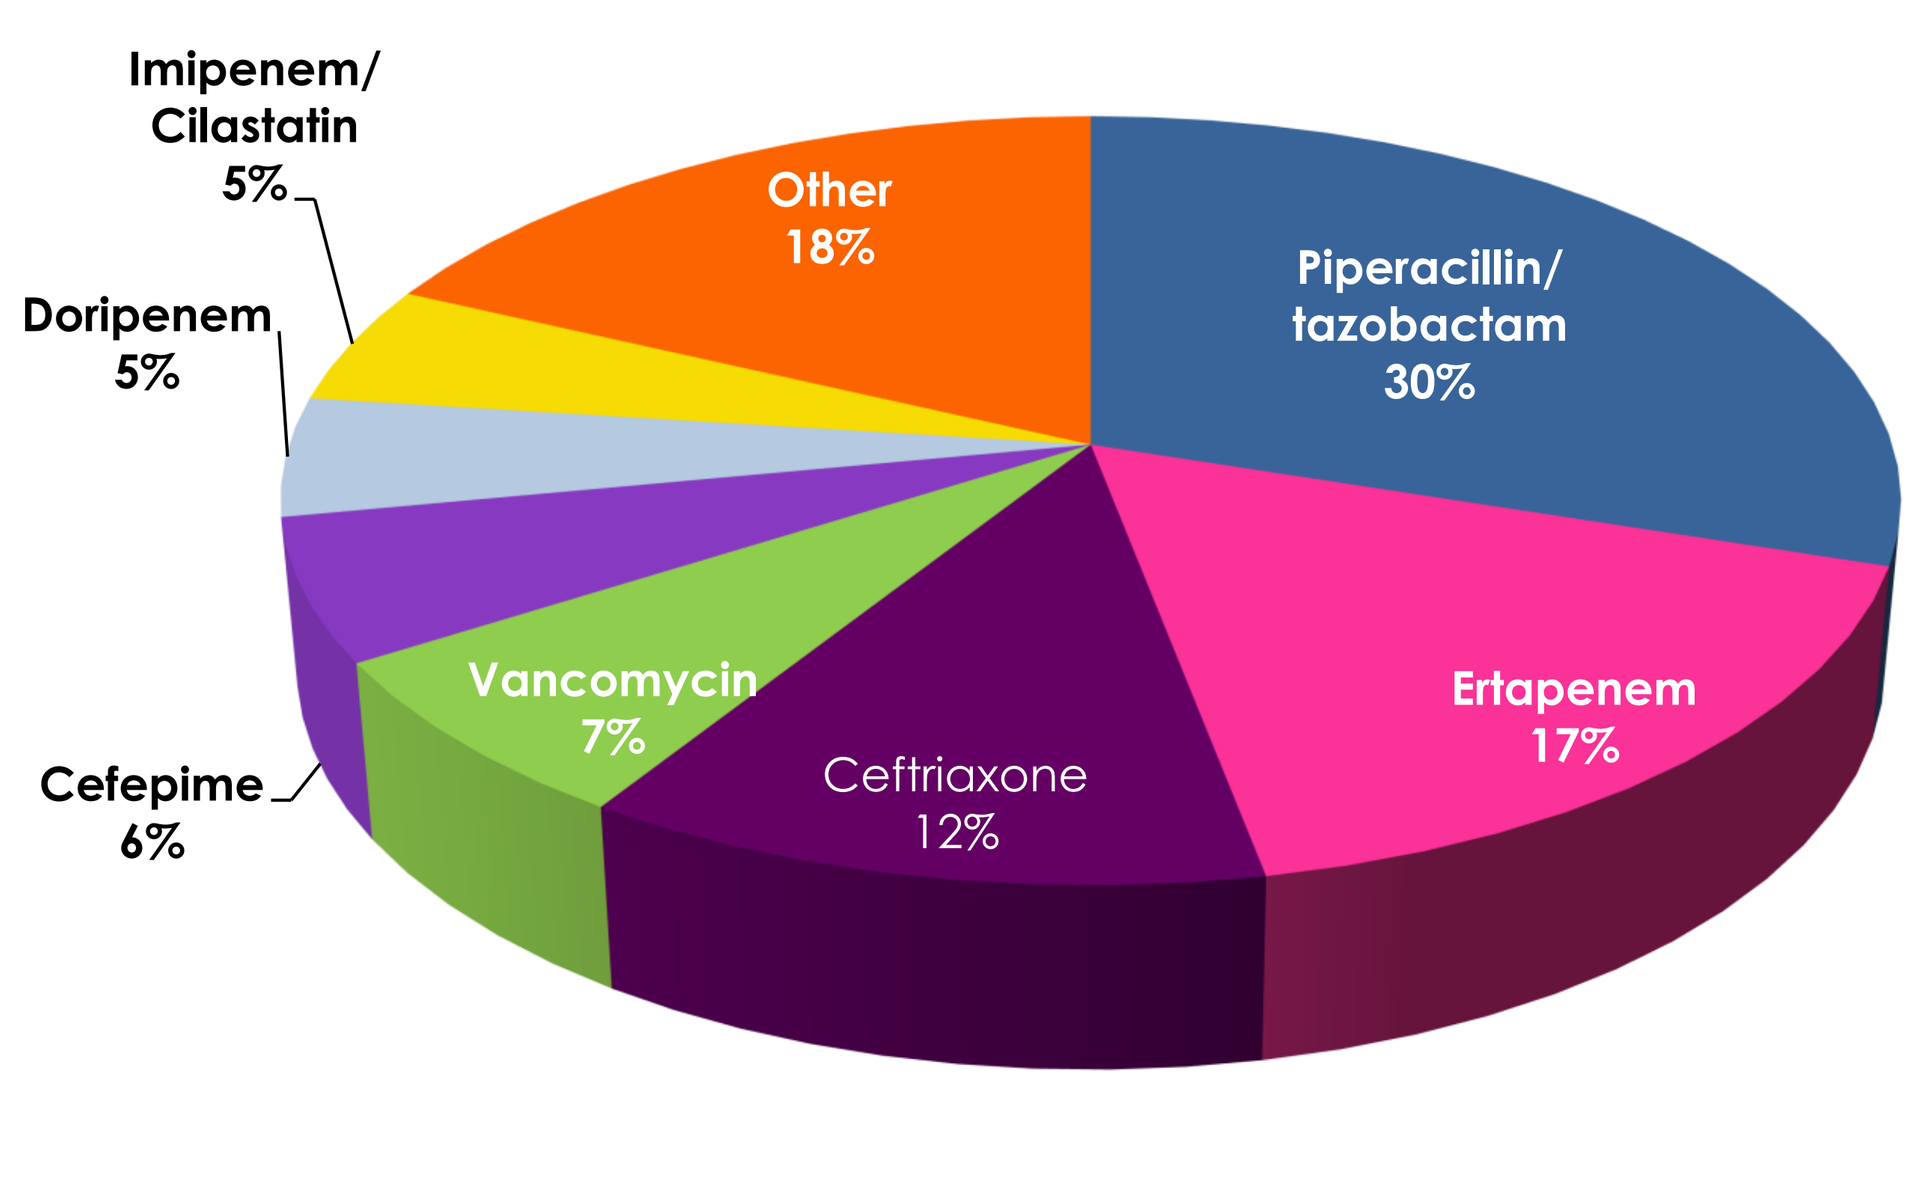
Imipenem/ Cilastatin 5%
Doripenem 5%
Cefepime 6%
Other 18%
Piperacillin/ tazobactam 30%
Ertapenem 17%
Ceftriaxone 12%
Vancomycin 7%

- 142 drugs were used in 120 patients. Twenty patients had 2 or more agent therapy regimens.
- Other IV drugs used in order of frequency included: daptomycin, cefazolin, micafungin, levofloxacin, telavancin, tigecycline, ampicillin/sulbactam, aztreonam, metronidazole, nafcillin and penicillin G.

Figure 4. Mean Duration of Therapy

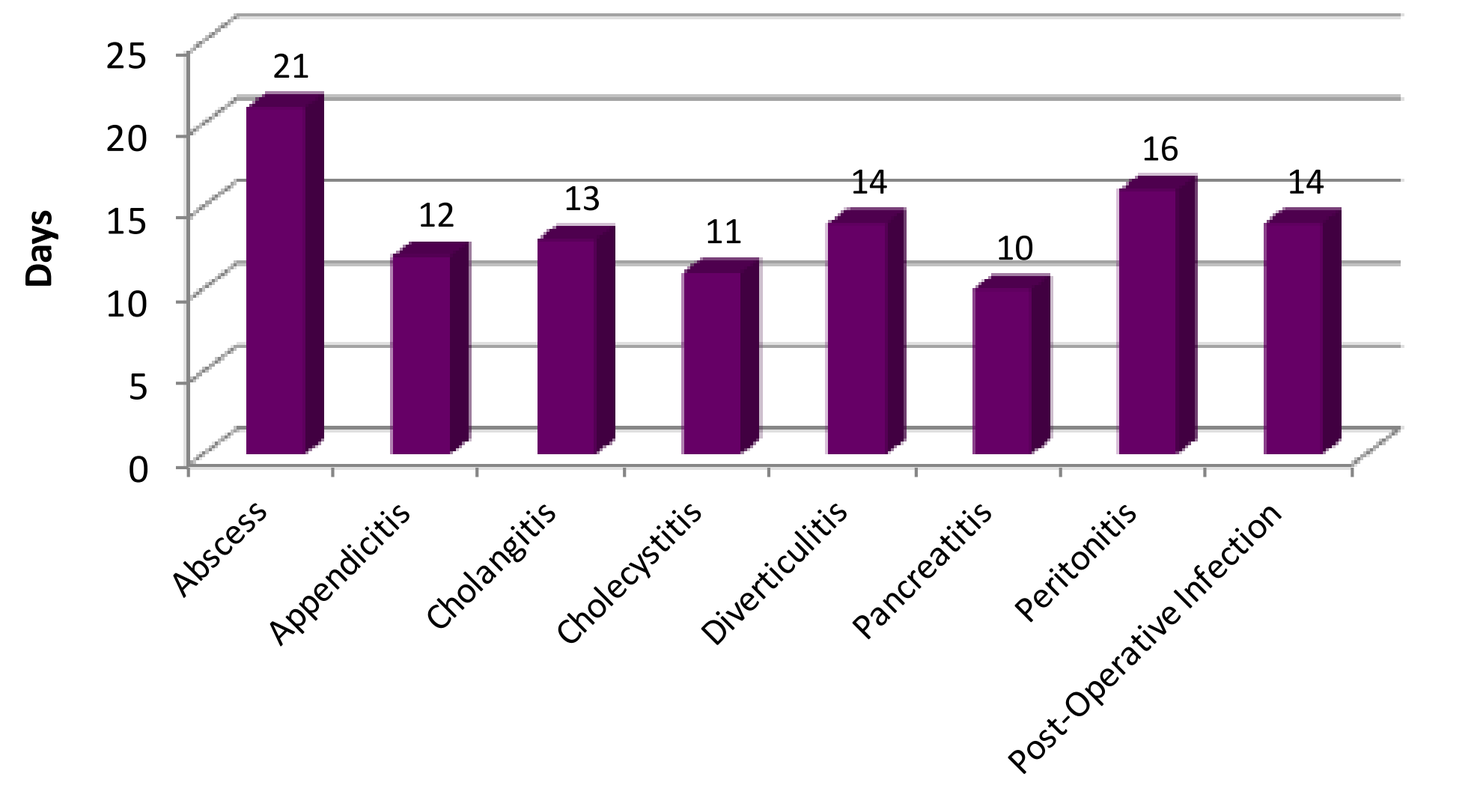
Days
25
20
15
10
5
0
21
12
13
11
14
10
16
14
Abscess
Appendicitis
Cholangitis
Cholecystitis
Diverticulitis
Pancreatitis
Peritonitis
Post-Operative Infection

- The average OPAT LOT for all diagnoses was 15 days.

Table 3. Drug-related Adverse Events

Adverse Events	No. (%)	Suspected Antibiotic
Aches/Soreness in legs	1 (0.8%)	Piperacillin/Tazobactam
Diarrhea	1 (0.8%)	Cefepime
Elevated LFTs	1 (0.8%)	Levofloxacin/Vancomycin
Fever	1 (0.8%)	Vancomycin or Piperacillin/Tazobactam
Headaches	1 (0.8%)	Vancomycin
Hemolytic Anemia*	1 (0.8%)	Piperacillin/Tazobactam
Increased SCr	1 (0.8%)	Piperacillin/Tazobactam
Nausea	4 (3%)	Imipenem/Cilastatin, Ampicillin/Sulbactam, Ciprofloxacin, Cefazolin
Rash	4 (3%)	Ceftriaxone, Vancomycin, Piperacillin/Tazobactam, Imipenem/Cilastatin
Vomiting	1 (<1%)	Cefazolin

* Serious adverse event still unresolved at end of IVAB therapy.

Clinical Outcomes

Figure 5. Overall Outcomes

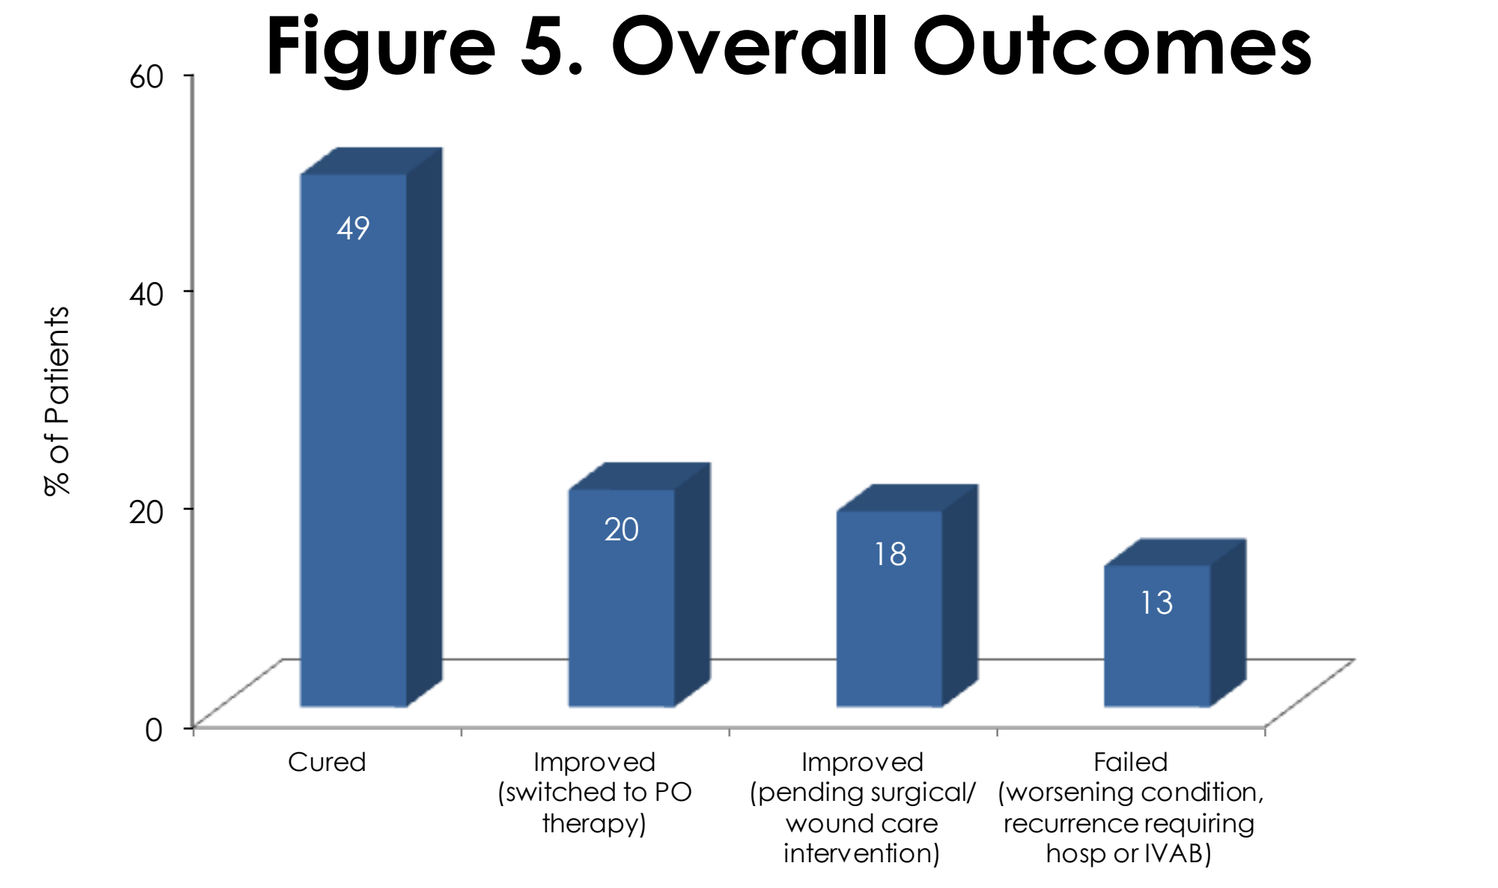
% of Patients
60
40
20
0
49
20
18
13
Cured
Improved (switched to PO therapy)
Improved (pending surgical/ wound care intervention)
Failed (worsening condition, recurrence requiring hosp or IVAB)

Figure 6. Outcomes by Diagnosis

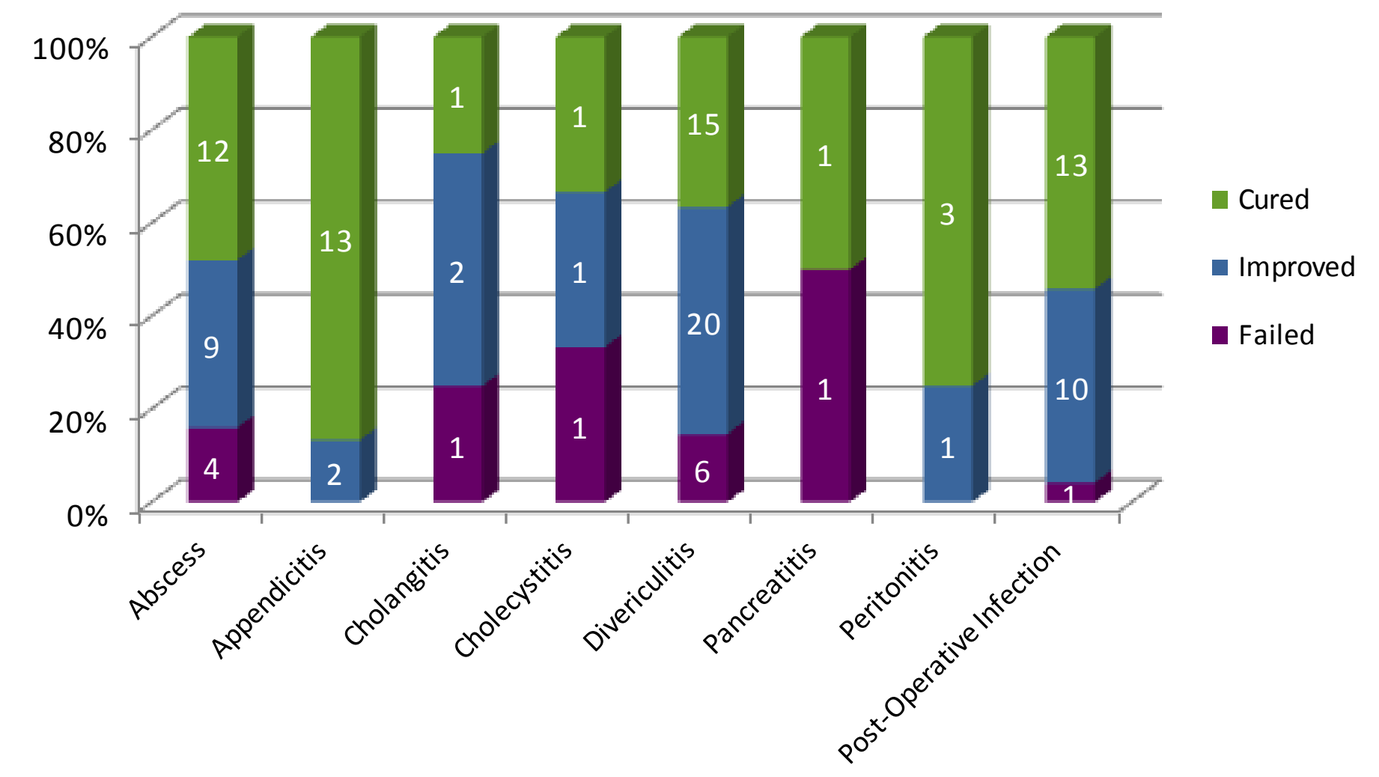
100%
80%
60%
40%
20%
0%
Abscess
Appendicitis
Cholangitis
Cholecystitis
Diverticulitis
Pancreatitis
Peritonitis
Post-Operative Infection
Cured
Improved
Failed

- Overall, 59 (49%) pts were cured, and 45 (38%) improved for overall clinical success of 87%.
- 16 (13%) pts failed, 6 due to worsening condition and 10 who had recurrences within 3 months following initial response.
- No catheter infections occurred in any patients during the therapy.

Discussion

- 120 Intra-abdominal infections were treated over 6-months in 10 POICs nationally.
- Patients had complicated infections with 87% requiring therapy post-hospital discharge.
- Patients had an even distribution between male and female, mean age of 52 and almost all had co-morbidities, 45% with 2 or more.
- Patients were treated for a wide range of IAI, most commonly diverticulitis (34%) and abscesses (22%).
- Infecting pathogens were consistent with reported organisms for IAI, with the addition of fungal organisms and *Mycobacterium chelonae*. 49% of patients had polymicrobial pathogens. 61% of gram negative organisms were MDR.
- The most commonly prescribed treatments for IAI in our patient population were piperacillin/tazobactam (30%), ertapenem (17%), and ceftriaxone (12%).
- Adverse event rates were low (13%) and all mild except for one case of hemolytic anemia with piperacillin/tazobactam.
- 87% of patients were treated successfully with disease recurrence in 16 pts (13%).

Conclusion

- Outpatient management of acute IA infections is safe and effective with the ability to substantially reduce length of hospital stay and costs.
- A variety of antibiotics were used successfully in the POIC setting.
- Further study is indicated with long-term follow-up.

References

1. Solomkin JS, Mazuski JE, Bradley JS, et al. Diagnosis and management of complicated intra-abdominal infection in adults and children: guidelines by the surgical infection society and the infectious diseases society of America. Clin Inf Dis 2010; 50: 133-164.
2. Center for Drug Evaluation and Research. Guidance for Industry Complicated Intra-Abdominal Infections: Developing Drugs for Treatment. U.S. Department of Health and Human Services: Food and Drug Administration 2012:1-23.
3. Sartelli M, Viale P, Catena F, et al. 2013 WSES Guidelines for Management of Intra-abdminal Infections. World Journal of Emergency Surgery 2013;8(3):1-29.

IDWeek 2013

October 2-6, 2013, San Francisco, CA