IDWeek™ 2023
#685

Atlanta ID Group

Early Experience with a Simple Administration of a Novel Fecal Microbiome Replacement for Prevention of Recurrent *Clostridioides difficile*

Richard L. Hengel MD[1]; Sujatha Krishan, MD[2]; Jonathan A. Rosenberg, MD[3]; Timothy E. Ritter, MD[4]; Kathy A. Baker Ph.D., APRN[5]; Lucinda J. Van Anglen, BS, PharmD[6]; Amy Guo, PhD[7]; Mielad Moosapanah, PharmD[7]; Min Yang, MD, PhD[8]; Kevin W. Garey, PharmD[9]

[1]Atlanta ID Group, Atlanta, GA; [2]DFW Infectious Disease, Dallas, TX; [3] GI Alliance, Highland Park, IL; [4]GI Alliance, Southlake, TX; [5]Harris College of Nursing and Health Sciences, Texas Christian University; [6]Healix Infusion Therapy, Sugar Land, TX; [7]Ferring Pharmaceuticals; [8]Analysis Group, Inc., Boston, MA; [9]University of Houston College of Pharmacy, Houston, TX

Introduction

Clostridioides difficile infection (CDI) is a global healthcare concern, causing between 15,000–30,000 deaths and healthcare costs exceeding $4.8 billion annually [1]. Even with appropriate treatment of CDI with standard-of-care (SoC) antibiotic therapy, first recurrences continue in up to 35% of patients following an initial episode [2-5]. While treating the CDI, SoC antibiotics disrupt the gut microbiota, increasing the risk for further recurrent CDI (rCDI). Microbial restoration with live biotherapeutic products may restore the diversity and composition of the gut microbiota to decrease the likelihood of rCDI.

Fecal Microbiota, live-jslm (RBL) is a rectally administered, pre-packaged, live biotherapeutic approved in November 2022 for the prevention of recurrence of *Clostridioides difficile* infection (rCDI) in adults [6]. Clinical trial data indicates success in prevention of recurrence [7,8] for RBL, the first FDA-approved microbiota product. Study is warranted of whether RBL, a novel therapy in preventing rCDI, may pose challenges in routine clinical administration, particularly with Infectious Disease (ID) or other specialties not typically providing rectally administered therapies.

Objectives

The objective of this study is to develop and report implementation of a simple administration protocol of RBL for rCDI prevention in routine clinical practice.

Methods

Study Design: Retrospective, multicenter cohort study conducted in physician office infusion centers (POICs) in the United States.

Patient Population: All patients who received at least one dose of RBL from February 2023 through May 2023, followed by pooled data of patients receiving RBL from May 2023 through August 2023.

Protocol: A multi-disciplinary protocol was developed for provision of RBL through POICs. Development included the following:

- Protocol designed by Nursing, Pharmacy, Business Office (BO), Operations, and Purchasing
- Certificate of Medical Necessity (CMN) for insurance approval
- Order set
- Guidelines for use:
 - Referral and order management
 - Notification to BO and Clinical Team of a new urgent (STAT) order
 - Insurance approval and submission of co-pay assistance, if applicable
 - Scheduling of patient with coordination of antibiotic discontinuation
 - Acquisition of RBL
 - Pre-appointment confirmation
 - Appointment procedures with preparation and administration

Data collection: Electronic medical records (EMR), administration records and internal databases were queried, and the following data collected:

- Patient demographics, including payor detail
- History of present illness (HPI) for current and past episodes of CDI
- CDI stool test results and antimicrobial therapy
- Time from order to treatment
- Nursing assessment for RBL administration
- Adverse effects during the procedure

Data Analysis: Continuous and categorical variables were reported using mean ±standard deviation (SD) or median with interquartile range (IQR) and frequency (percentage), respectively.

Study Cohort

Table 1. Patient Characteristics

- 25 patients from 12 POICs in 8 states have received RBL through 8/31/2023.
- 11 patients have been treated in Infectious Disease POICs and 14 in GI POICs.

Characteristic	Results (N=6) through May 2023	Results (N=25) through Aug 2023
Age, years (mean±SD)	76±11	69±17
Age ≥65 years	5 (83%)	17 (68%)
Female	4 (67%)	17 (68%)
CDI episodes, including current, median (IQR)	4 (3-5)	4 (3-4)
SoC Antibiotic for the current episode of CDI		
Vancomycin	3 (50%)	12 (48%)
Fidaxomicin	2 (33%)	8 (32%)
Fidaxomicin and vancomycin	1 (17%)	4 (16%)
Other*	0	2 (8%)
Health Insurance		
Commercial	1 (17%)	9 (36%)
Medicare, Traditional	3 (50%)	9 (36%)
Medicare, Advantage	2 (33%)	7 (28%)

*vancomycin + metronidazole (n=1), unknown antibiotic (n=1); Data are presented as no. (%) unless otherwise indicated.

Figure 1. Certificate of Medical Necessity

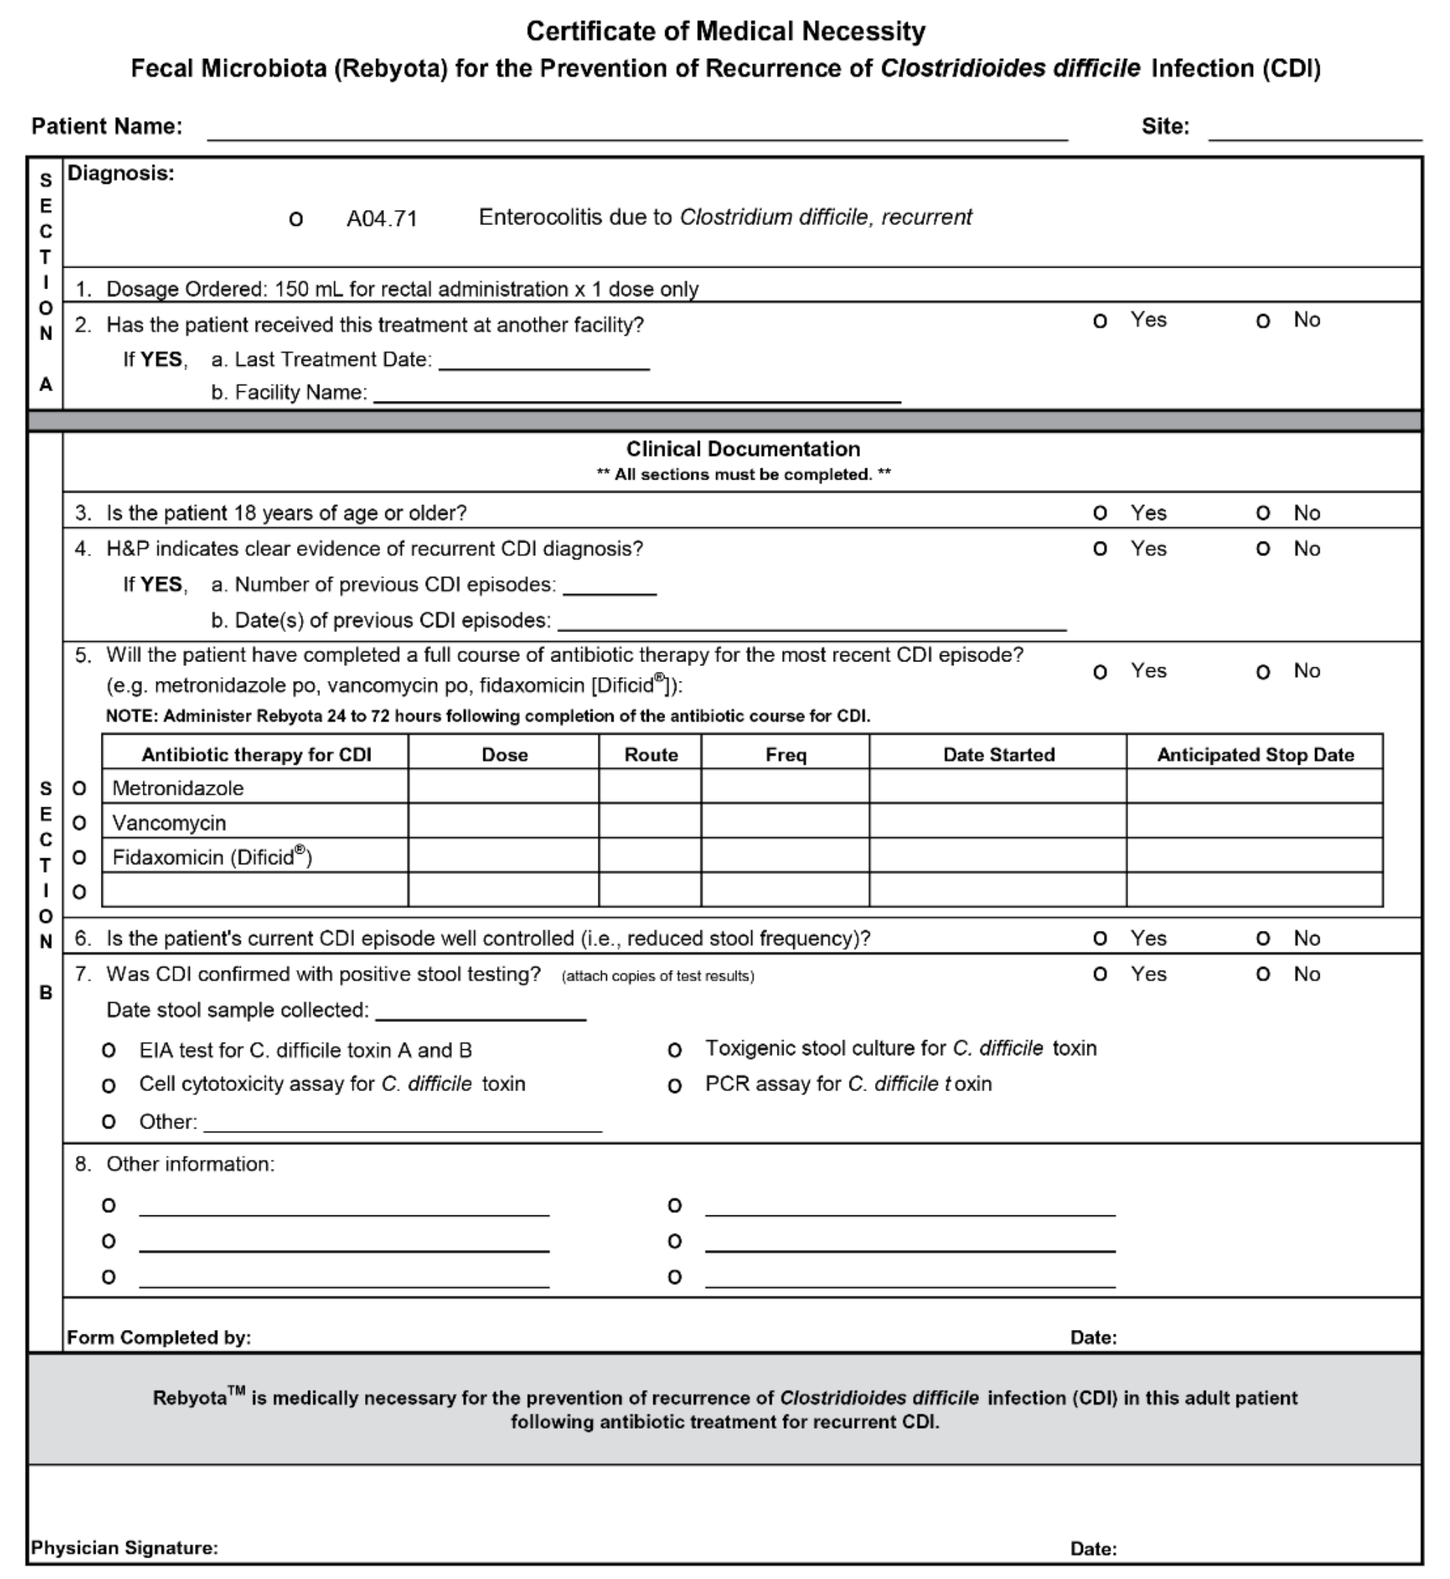

Certificate of Medical Necessity
Fecal Microbiota (Rebyota) for the Prevention of Recurrence of *Clostridioides difficile* Infection (CDI)

Patient Name: ______ Site: ______

SECTION A
Diagnosis:
o A04.71 Enterocolitis due to *Clostridium difficile, recurrent*
1. Dosage Ordered: 150 mL for rectal administration x 1 dose only
2. Has the patient received this treatment at another facility? o Yes o No
If YES, a. Last Treatment Date: ______
b. Facility Name: ______

SECTION B
Clinical Documentation
** All sections must be completed. **
3. Is the patient 18 years of age or older? o Yes o No
4. H&P indicates clear evidence of recurrent CDI diagnosis? o Yes o No
If YES, a. Number of previous CDI episodes: ______
b. Date(s) of previous CDI episodes: ______
5. Will the patient have completed a full course of antibiotic therapy for the most recent CDI episode? (e.g. metronidazole po, vancomycin po, fidaxomicin [Dificid®]): o Yes o No
NOTE: Administer Rebyota 24 to 72 hours following completion of the antibiotic course for CDI.

	Antibiotic therapy for CDI	Dose	Route	Freq	Date Started	Anticipated Stop Date
o	Metronidazole					
o	Vancomycin					
o	Fidaxomicin (Dificid®)					
o						

6. Is the patient's current CDI episode well controlled (i.e., reduced stool frequency)? o Yes o No
7. Was CDI confirmed with positive stool testing? (attach copies of test results) o Yes o No
Date stool sample collected: ______
o EIA test for C. difficile toxin A and B o Toxigenic stool culture for *C. difficile* toxin
o Cell cytotoxicity assay for *C. difficile* toxin o PCR assay for *C. difficile* toxin
o Other: ______
8. Other information:
o ______ o ______
o ______ o ______
o ______ o ______

Form Completed by: ______ Date: ______

Rebyota™ is medically necessary for the prevention of recurrence of *Clostridioides difficile* infection (CDI) in this adult patient following antibiotic treatment for recurrent CDI.

Physician Signature: ______ Date: ______

Table 2. Treatment Criteria

RBL Protocol Treatment Criteria
Primary Diagnosis: Enterocolitis due to Clostridium difficile, recurrent (ICD-10 Code: A04.71)
Patient Age (required to be ≥18 years)
Completion of Certificate of Medical Necessity, including the following:
History and Physical
Clear indication of current episode of recurrent CDI
Prior history of CDI, including number of episodes and dates
Positive stool test(s) for current episode, with type of test and dates
Antibiotic therapy for the current CDI episode
Antibiotic regimen and planned stop date (to plan for RBL administration)
Confirmation of control of symptoms with the current episode

Logistical Flow

Figure 2. Process Flow

Order written and submitted in EMR for RBL
↓
Clinician compiles clinical notes and lab results to document medical necessity of RBL
(must include HPI, positive stool test results, SoC antibiotic treatment)
↓
Benefits verified STAT and case reviewed by clinical pharmacist
↓
Authorization obtained STAT from insurance company as required.
↓
Appointment scheduled upon insurance approval
(approx. 48 hrs. following completion of SoC)*
↓
RBL is ordered 2-4 days prior to appointment
↓
RBL administered to patient

*RBL typically scheduled for 48 hrs. post SoC to allow adequate time for drug delivery and thawing.

Table 3. Clinical and Administration Protocol

RBL Clinical Protocol
Appointment Coordination
Ensure SoC antibiotic therapy has been discontinued 24-72 hours prior to RBL administration.
Order RBL 2-4 days prior to the appointment for overnight delivery Tuesday through Friday.
Store RBL in the refrigerator immediately upon receipt. (Outside of a -70°C environment, RBL is stable for 5 days refrigerated.)
Pre-appointment Confirmation (24 hours prior)
Confirm patient does not have active diarrhea or continuing CDI symptoms.
Confirm the SoC antibiotic has been or will be discontinued within 24-72 hours of the appointment.
Thaw RBL in the refrigerator for 24 hours prior to the appointment.
Remind patient to evacuate bowel prior the appointment, but not to perform any bowel prep.
RBL Administration
Greet patient and room them in a private room with an exam table.
Have patient evacuate bladder and bowels if not done prior to appointment.
Don proper PPE and follow infection control guidelines for Clostridioides difficile.
Retrieve RBL from the refrigerator just prior to administration.
Place underpad on the exam table and position patient in a position as noted in Figure 2.
Prepare RBL for administration with attachment of provided tubing and apply lubricant to tip.
Administer single dose of RBL 150 mL immediately via gravity flow over 5 minutes.
Keep patient in the same position and observe for 15 minutes to minimize cramping.
Allow patient to leave the clinic (may use bathroom prior to leaving if needed).

Abbreviations: PPE, personal protective equipment

Administration

Figure 3. Administration Diagrams

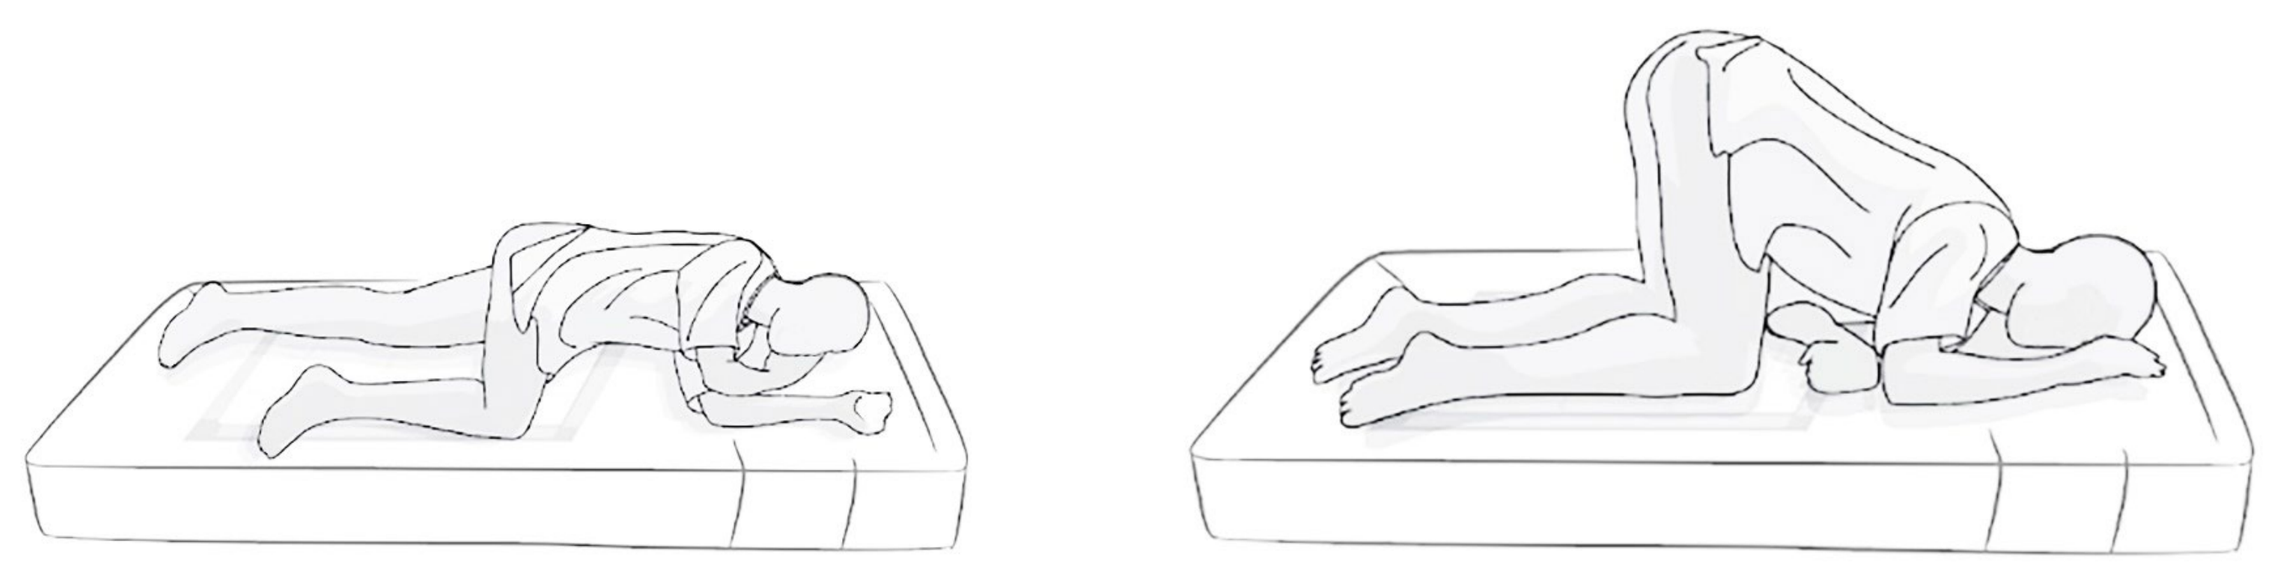

Left-side position: Lie on left side with knee bent and arms resting comfortably.

Knee-chest position: Kneel, then lower head and chest forward until left side of face is resting on surface with left arm folded comfortably.

Figure 4. RBL Order

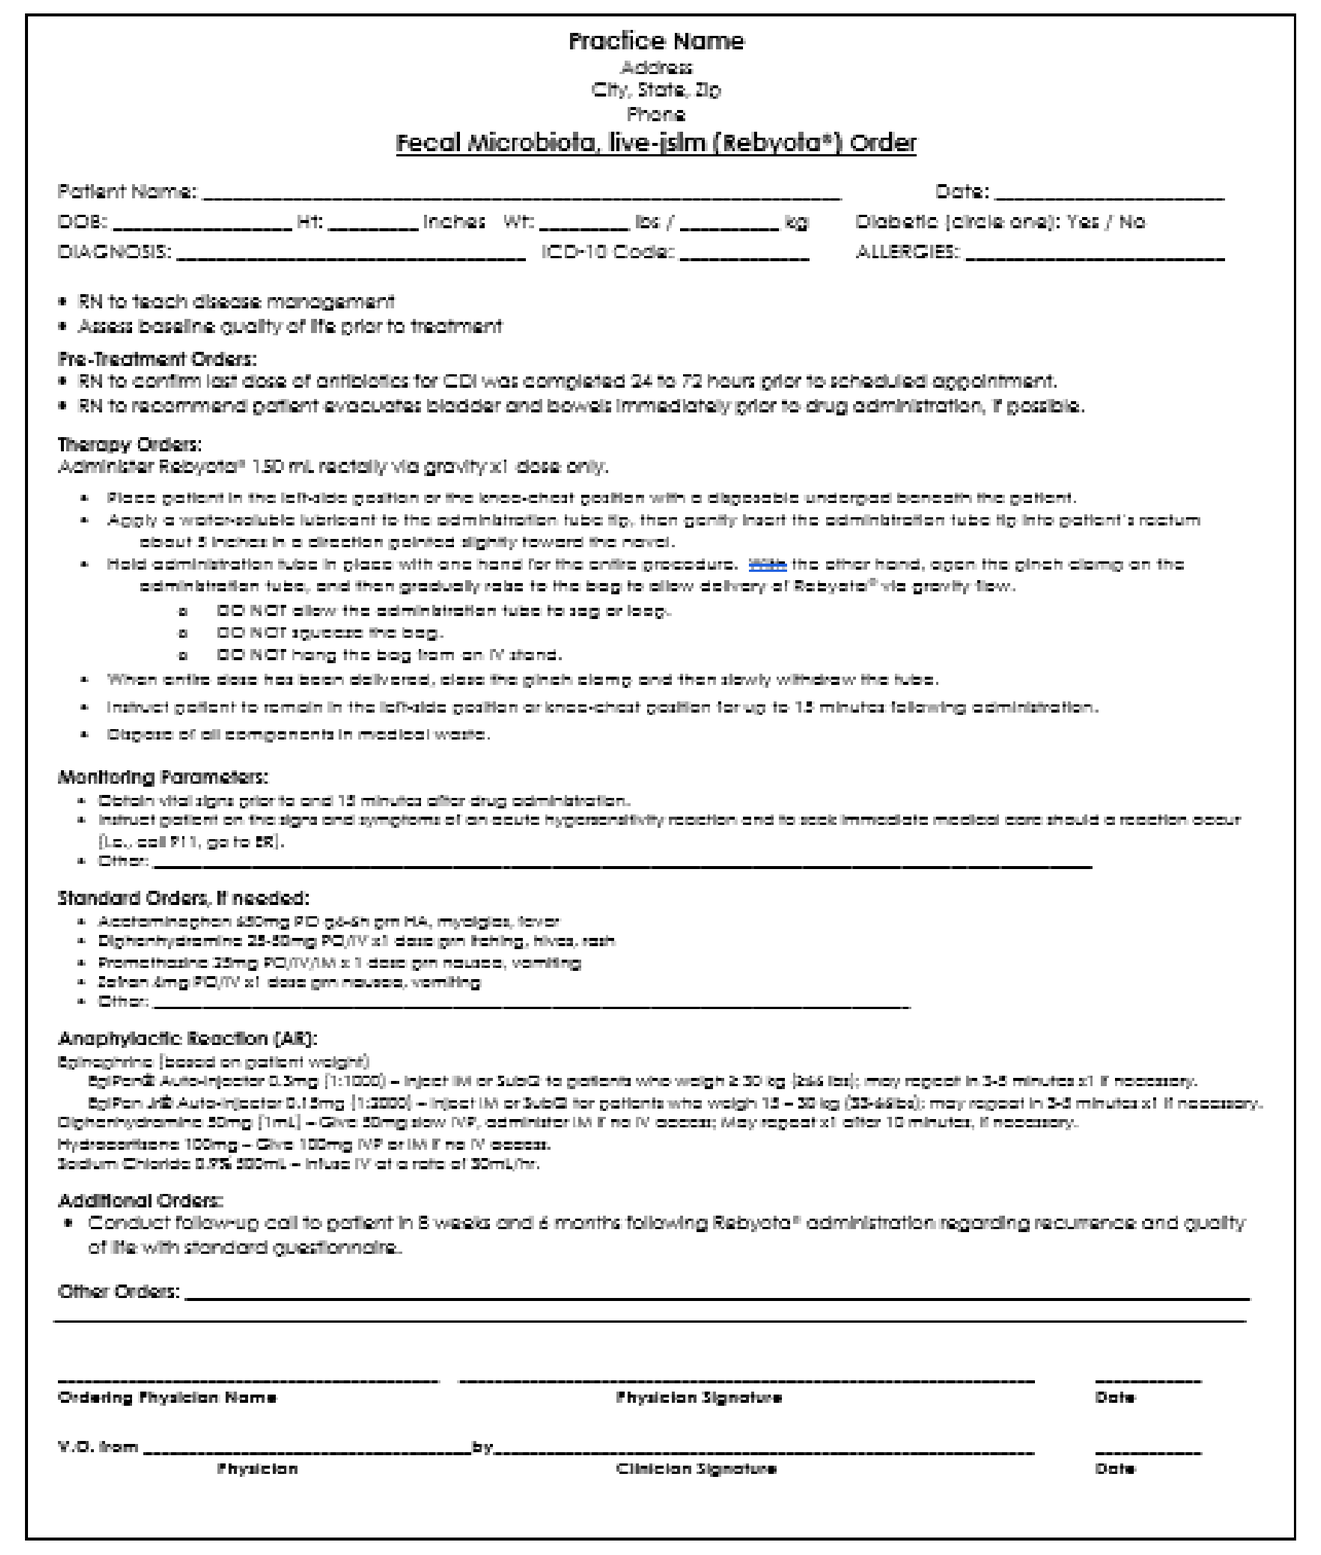

Practice Name
Address
City, State, Zip
Phone
Fecal Microbiota, live-jslm (Rebyota®) Order

Patient Name: ______ Date: ______
DOB: ______ Ht: ______ inches Wt: ______ lbs / ______ kg Diabetic (circle one): Yes / No
DIAGNOSIS: ______ ICD-10 Code: ______ ALLERGIES: ______

- RN to teach disease management
- Assess baseline quality of life prior to treatment

Pre-Treatment Orders:
- RN to confirm last dose of antibiotics for CDI was completed 24 to 72 hours prior to scheduled appointment.
- RN to recommend patient evacuates bladder and bowels immediately prior to drug administration, if possible.

Therapy Orders:
Administer Rebyota® 150 mL rectally via gravity x1 dose only.

Monitoring Parameters:

Standard Orders, if needed:

Anaphylactic Reaction (AR):

Additional Orders:
- Conduct follow-up call to patient in 8 weeks and 6 months following Rebyota® administration regarding recurrence and quality of life with standard questionnaire.

Other Orders: ______

Ordering Physician Name ______ Physician Signature ______ Date
V.O. from ______ by ______ Physician Clinician Signature Date

Table 4. Administration Outcomes

Parameter	Cohort through May 2023: No. of pts (N=6)*	Cohort through May 2023: Result	Cohort through Aug 2023*: No. of pts (N=25)*	Cohort through Aug 2023*: Result
Time from order to RBL administration, median days (IQR)	6	22 (18-26)	25	19 (17-30)
Instillation time via gravity flow, minutes, median (IQR)	1	18	12	6 (5-12)
Observation time, minutes	3	15	17	15
Administration position	6	-	16	-
Left lateral	-	6	-	15
Right lateral	-	0	-	1
Knee-Chest	-	0	-	0
Healthcare provider administering	6	-	22	-
Registered Nurse	-	1	-	16
Licensed Vocational Nurse	-	3	-	3
Nurse Practitioner	-	1	-	2
Physician	-	1	-	1
Adverse effects reported during the procedure	6	0	22	0
Reports of rectal leakage	6	0	22	0
Patients with bowel movement after the procedure	6	2	22	4

*Numbers noted for each parameter reflect those reported.

Discussion

This multicenter study was a development and analysis of a simple protocol for administration of RBL, a rectally administered, pre-packaged, live biotherapeutic for treatment of 2022 for the prevention of rCDI in adults.

- 25 patients received RBL following development and initiation of a protocol for use in 12 physician office infusion centers.
- Patients were older (69±17 years) with the majority (64%) Medicare or Medicare Advantage recipients. Median number of episodes, including current was 4 and vancomycin was the most used standard of care antibiotic for treatment of CDI.
- Protocol key success factors were:
 - Rapid compilation of required records for insurance approval with completion of a Certificate of Medical Necessity and STAT submission of the order. Median approval was 19 days from order to treatment.
 - Training for the nurses of the protocol and procedures for management, drug ordering, patient communication, and administration
 - Prompt order placement and receipt of RBL to meet appointments.
- This resulted in successful management of the patient. Once insurance was approved the patient was scheduled with confirmation of completion of SoC, RBL was ordered for arrival 2-4 days prior to administration.
- Administration was completed in 6 minutes with 15 minutes of observation. No leakage or adverse events occurred.
- All patients adhered to appointments and received RBL as planned.

Conclusion

- **Development of a multi-disciplinary protocol was critical in facilitation of the novel RBL therapy in physician office infusion centers, particularly with rectal administration.**
- **Time from order to treatment was 2.7 weeks, correlating well with completion of antibiotic therapy for treatment of the CDI episode.**
- **Overall patient visit time and administration of RBL was brief safe, and simple.**

References

1. McDonald LC, et al. Clin Infect Dis. 2018; 66:e1-e48.
2. Johnson S, et al. Clin Infect Dis. 2021;73(5):e1029-44.
3. Guh A, et al. N Engl J Med. 2020; 382:1320-1330
4. Cornely OA, et al. Clin Infect Dis. 2012; 55:S154-61.
5. Ma GK, et al. Ann Intern Med. 2017; 167:152-8.
6. Rebyota (fecal microbiota, live - jslm) [package insert]. Roseville, MN: Ferring Pharmaceuticals; 2022.
7. Dubberke ER, et al. Infect Dis Ther. 2023; 12:703–710.
8. Khanna S, et al. Drugs. 2022; 82:1527–1538.

IDWeek™ 2023 Oct 11-15, 2023; Boston, MA